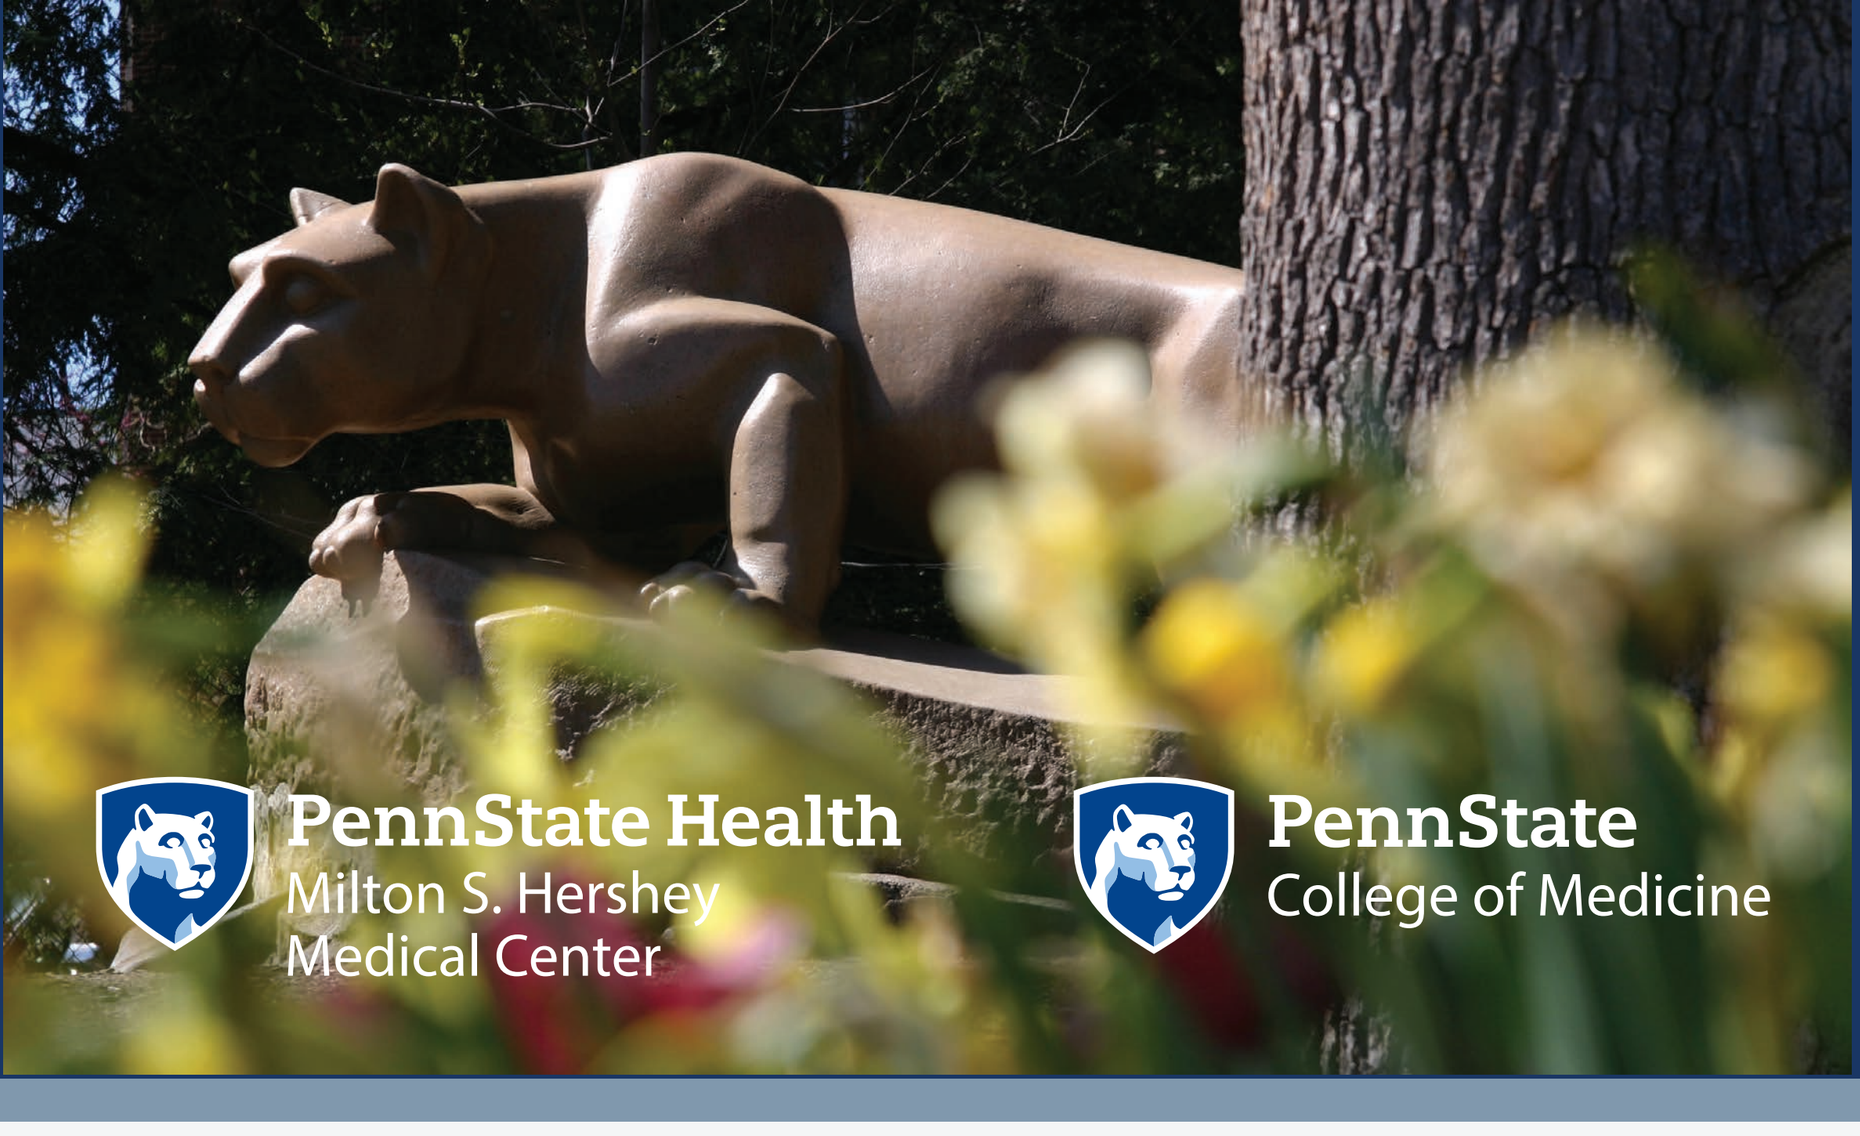
PennState Health
Milton S. Hershey
Medical Center
PennState
College of Medicine

Adrenalectomy: Shouldn’t Urologists Be Doing More?

MP03

Jay G. Fuletra[1], Amber Schilling[2], Christopher Hollenbeak[3], Jay D. Raman[1]

[1]Department of Surgery, Division of Urology, Penn State Health Milton S. Hershey Medical Center, Hershey, PA
[2]Penn State College of Medicine, Hershey, PA
[3]Penn State University, University Park, PA

Introduction:

- Adrenalectomy is an operation increasingly performed by general surgeons.
- Urologists, however, are extremely familiar with the surgical anatomy of the retroperitoneum.
- It is unclear if differences exist in perioperative outcomes based on surgical training background.
- If comparable outcomes are observed, urologists should maintain adrenalectomies in surgical repertoire.

Objective:

- To investigate if differences in perioperative outcomes exist based on surgical specialty performing the adrenalectomy procedure.

Methods:

- National Surgical Quality Improvement Project (NSQIP) Participant Use File (PUF) queried to extract all adrenalectomy cases performed during the years 2011-2015.
- CPT codes: 60540 (open) and 60650 (laparoscopic)
 - Patients with any concurrent visceral resections excluded
- Data stratified by surgical specialty (urology vs. general surgery)
- Outcomes of interest
 - post-surgical complications
 - reoperations
 - hospital length of stay (LOS)
 - 30-day readmission
 - mortality

Results:

- 3,346 adrenalectomy patients included
 - 3,012 (90%) by general surgeon
 - 334 (10%) by urologist
- Patients operated on by urologists **(Table)** were:
 - older (p=0.013)
 - non-black race (p=0.021)
 - male gender (p<0.0001)
 - more likely to undergo an open operation (p=0.013)
 - had longer operative duration (p=0.0059)
- On univariate analysis **(Table)**, no differences in:
 - Number (p=0.76) or type of post-surgical complications
 - LOS (p=0.29)
 - Rate of reoperation (p=0.37)
 - 30-day readmission rate (p=0.98)
 - Perioperative mortality (p=0.59)
- Multivariate regression models confirmed no difference in urologic (vs. general surgeon) outcomes:
 - Any post-operative complication (HR 1.13, p=0.57)
 - Need for reoperation (HR 0.46, p=0.29)
 - 30-day readmission rate (HR 0.98, p=0.94)
 - Perioperative mortality (HR 0.66; p=0.69)

Conclusion:

- Surgical specialty did not impact outcomes for patients undergoing adrenalectomy.
- Given the absence of difference, urologists should continue to include adrenalectomy procedures within their practice when feasible.

Acknowledgements:

- We gratefully acknowledge the Keith and Lynda Harring Fund for Urology Research.

Table:

Demographic characteristics and selected outcomes for the adrenalectomy cohort, stratified by surgical specialty performing the procedure.

	Surgical Specialty			
Variable	Overall Cohort *(N=3346)*	General *(N=3012)*	Urology *(N=334)*	P-Value
Age (mean, years)	53.7	53.5	55.8	0.013
Race				0.0010
White	66.7%	66.9%	64.7%	0.42
Black	13.9%	14.4%	9.9%	0.021
Hispanic	6.3%	6.2%	6.6%	0.77
Other	4.5%	4.5%	4.2%	0.90
Sex				<0.0001
Male	42.0%	40.6%	54.5%	
Female	58.0%	59.4%	45.5%	
Body Mass Index (mean, kg/m2)	30.0	30.0	30.1	0.68
Comorbidities / Pre-Surgical Risk Factors				
Diabetes	19.5%	19.9%	16.2%	0.13
Smoking	23.4%	23.6%	22.5%	0.50
Chronic Obstructive Pulmonary Disease	5.7%	5.8%	4.8%	0.51
Congestive Heart Failure	0.6%	0.6%	0.3%	0.47
Hypertension	70.5%	71.0%	66.2%	0.05
Acute Renal Failure or Dialysis	0.7%	0.8%	0.0%	0.096
Bleeding Disorder	2.6%	2.7%	2.4%	0.71
ASA Class	2.7	2.7	2.7	0.67
Post-Surgical Complications				
Surgical Site Infection	0.5%	0.5%	0.6%	0.84
Pneumonia	1.3%	1.4%	0.6%	0.19
Reintubation (unplanned)	0.9%	1.0%	0.3%	0.19
Renal Insufficiency or Acute Renal Failure	0.6%	0.6%	0.6%	0.97
Urinary Tract Infection	1.3%	1.3%	0.6%	0.23
Cardiac Complication	0.4%	0.3%	0.9%	0.093
Transfusion Intra-Op / Post-Op	5.8%	5.5%	8.4%	0.055
Blood Clot	0.9%	0.9%	1.2%	0.58
Pulmonary Embolism	0.6%	0.6%	0.9%	0.49
Deep Vein Thrombosis	0.5%	0.5%	0.6%	0.84
Sepsis or Septic Shock	1.0%	1.0%	0.3%	0.18
Surgical Approach				0.013
Open	16.2%	15.6%	20.1%	
Laparoscopic	83.8%	84.4%	79.9%	
Operative Time (mean, hours)	2.4	2.4	2.6	0.0059
Length of Stay (mean, days)	3.3	3.3	2.9	0.29
Reoperation	1.0%	1.1%	0.6%	0.37
Readmitted	5.7%	5.7%	5.4%	0.98
Died	0.48%	0.50%	0.30%	0.59