Abstract MP52-04

2-Year Follow-up of Radium-223 Re-treatment in an International, Open-Label, Phase 1/2 Study in Patients With Castration-Resistant Prostate Cancer and Bone Metastases

Neil Mariados,[1] Oliver Sartor,[2] Daniel Heinrich,[3] Maria José Méndez Vidal,[4] Daniel Keizman,[5] Camilla Thellenberg Karlsson,[6] Avivit Peer,[7] Giuseppe Procopio,[8] Stephen J. Frank,[9] Kalevi Pulkkanen,[10] Eli Rosenbaum,[11] Stefano Severi,[12] Jose Manuel Trigo Perez,[13] Lucia Trandafir,[14] Volker Wagner,[14] Rui Li,[15] Luke T. Nordquist[16]

[1]Associated Medical Professionals of New York, PLLC, Syracuse, NY, USA; [2]Tulane Cancer Center, New Orleans, LA, USA; [3]Akershus University Hospital, Lørenskog, Norway; [4]Maimonides Institute of Biomedical Research (IMIBIC), Reina Sofia Hospital, University of Cordoba, Cordoba, Spain; [5]Meir Medical Center, Kfar Saba, Israel; [6]Cancer Center Norrlands University, Umeå, Sweden; [7]Rambam Health Care Campus, Haifa, Israel; [8]Fondazione Istituto Nazionale Tumori Oncologia Medica Genitourinaria, Milan, Italy; [9]Hadassah Hebrew University Medical Center, Jerusalem, Israel; [10]Kuopio University Hospital, Kuopio, Finland; [11]Rabin Medical Center–Davidoff Center, Petah Tikva, Israel; [12]Romagnolo Scientific Institute for the Study and Care of Cancer—IRST IRCCS, Meldola, Italy; [13]Hospital Universitario Virgen de la Victoria, Málaga, Spain; [14]Bayer Pharma AG, Basel, Switzerland; [15]Bayer HealthCare Pharmaceuticals, Whippany, NJ, USA; [16]GU Research Network, LLC, Omaha, NE, USA

BACKGROUND

Radium-223 Dichloride (Radium-223)

- First targeted alpha therapy approved for patients with metastatic castration-resistant prostate cancer (mCRPC) (**Figure 1**)[1]
 - Shown to improve survival versus placebo in the phase 3 ALSYMPCA trial[2]
 - Well tolerated with low rates of myelosuppression[2]

Figure 1. Radium-223 Mechanism of Action

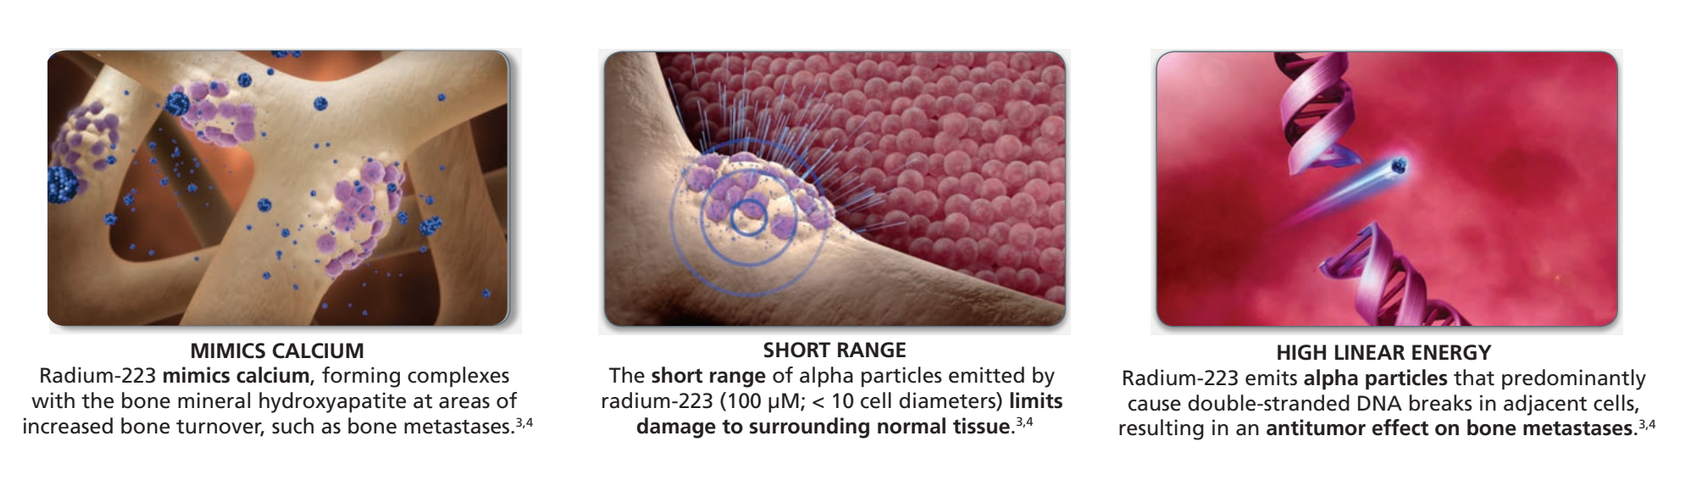
MIMICS CALCIUM
Radium-223 mimics calcium, forming complexes with the bone mineral hydroxyapatite at areas of increased bone turnover, such as bone metastases.3,4
SHORT RANGE
The short range of alpha particles emitted by radium-223 (100 µM; < 10 cell diameters) limits damage to surrounding normal tissue.3,4
HIGH LINEAR ENERGY
Radium-223 emits alpha particles that predominantly cause double-stranded DNA breaks in adjacent cells, resulting in an antitumor effect on bone metastases.3,4

- Efficacy and safety from ALSYMPCA are based on a dosing regimen of 1 injection (55 kBq/kg IV) every 4 weeks for a total of 6 injections[1]
- Early results of this international, open-label, phase 1/2 study (re-treatment study; NCT01934790) showed that re-treating patients with up to 6 additional radium-223 injections was well tolerated with favorable effects on disease progression[5]

RATIONALE AND OBJECTIVES

- Re-treatment with radium-223 may be well tolerated and provide further benefit to patients who received an initial course of 6 radium-223 injections
- Reported here are safety and efficacy results from a 2-year follow-up of the radium-223 re-treatment study (NCT01934790) in patients with bone-metastatic CRPC

REFERENCES

1. Xofigo (radium Ra 223 dichloride) injection, for intravenous use [package insert]. Wayne, NJ: Bayer Healthcare Pharmaceuticals Inc; May 2013.
2. Parker et al. *N Engl J Med*. 2013;369:213-223.
3. Kingsley et al. *Mol Cancer Ther*. 2007;6:2609-2617.
4. Suominen et al. *Clin Cancer Res*. 2017;23:4335-4346.
5. Sartor et al. *Ann Oncol*. 2017;28:2464-2471.

METHODS

- Study design is presented in **Figure 2**

Figure 2. Radium-223 Re-treatment Study Design

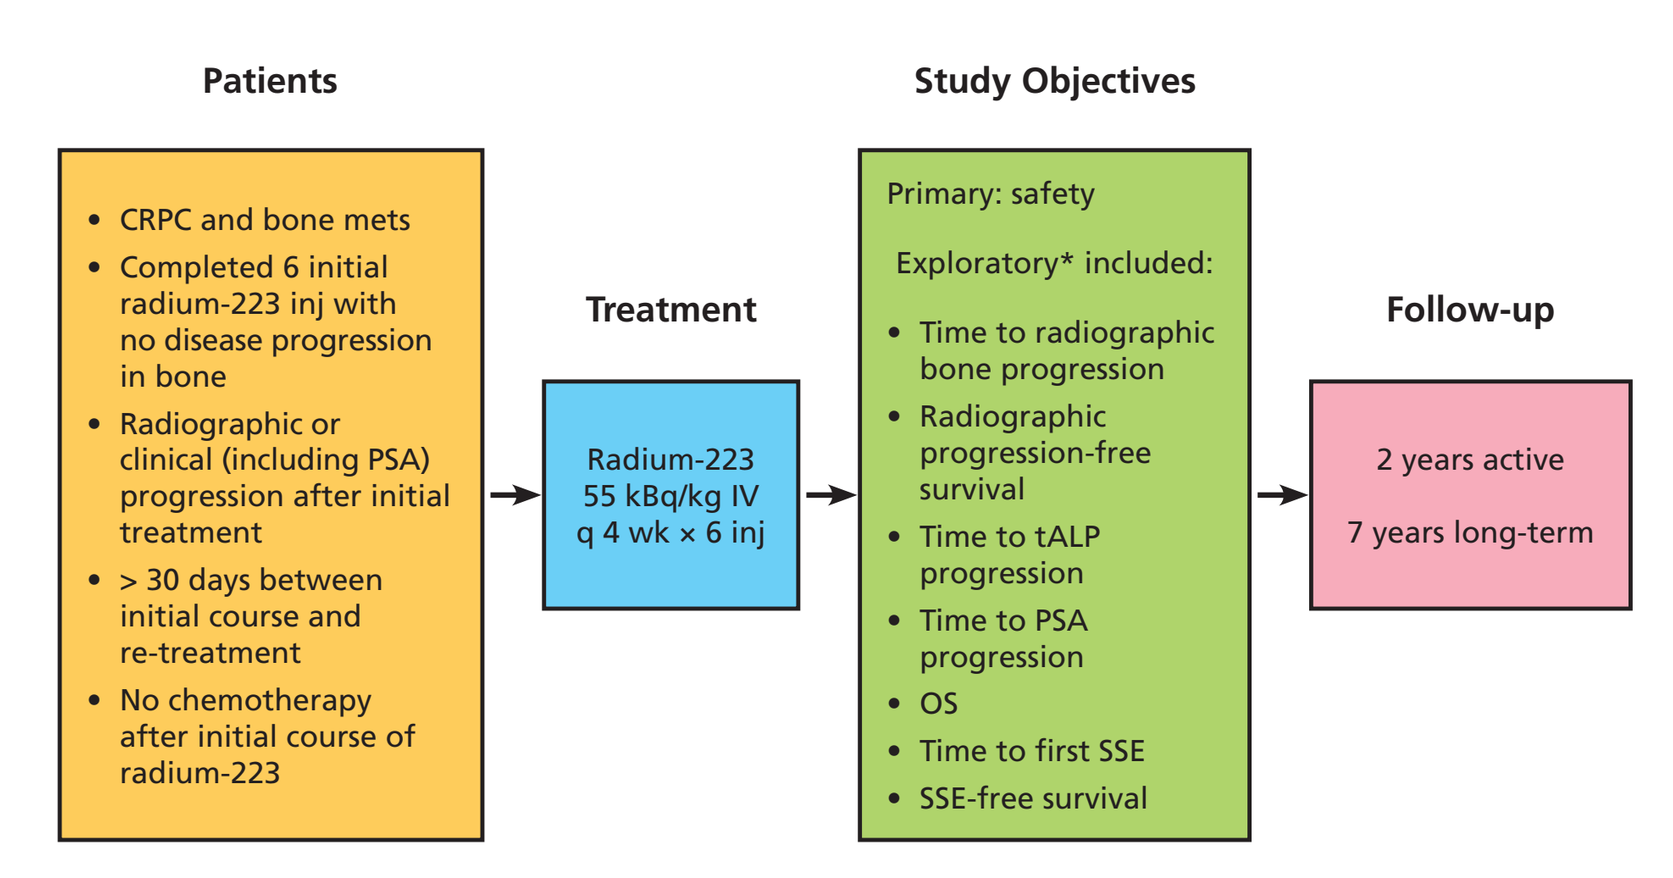
Patients
CRPC and bone mets
Completed 6 initial radium-223 inj with no disease progression in bone
Radiographic or clinical (including PSA) progression after initial treatment
> 30 days between initial course and re-treatment
No chemotherapy after initial course of radium-223
Treatment
Radium-223 55 kBq/kg IV q 4 wk × 6 inj
Study Objectives
Primary: safety
Exploratory* included:
Time to radiographic bone progression
Radiographic progression-free survival
Time to tALP progression
Time to PSA progression
OS
Time to first SSE
SSE-free survival
Follow-up
2 years active
7 years long-term

*Exploratory end points were calculated from start of radium-223 re-treatment.
CRPC = castration-resistant prostate cancer; inj = injections; mets = metastases; OS = overall survival; PSA = prostate-specific antigen; SSE = symptomatic skeletal event; tALP = total alkaline phosphatase.

Eligibility

- Eligible patients had CRPC and bone metastases and previously completed 6 initial radium-223 injections with
 - No progression in bone (according to Prostate Cancer Working Group 2 [PCWG2] criteria) during first treatment course
 - Radiographic progression or clinical progression (defined as 2 subsequent values showing prostate-specific antigen increase [≥ 2 ng/mL] or substantial worsening of pain) after initial treatment
 - Adequate hematologic values
- Patients were ineligible if they had radium-223–related serious or grade 3 or 4 adverse events (AEs), during or after initial treatment, that did not resolve or led to treatment discontinuation
- No chemotherapy after initial course of radium-223 was permitted
- No concomitant chemotherapy or systemic radioisotopes (or investigational drugs) were permitted
- Other concomitant agents were permitted at investigator's discretion

Study Dates

- 2-year active follow-up period began at final data cutoff (June 11, 2015); last patient last visit (LPLV) was April 12, 2017
 - Efficacy data reported here are cumulative from first patient first visit (FPFV) (December 22, 2013) to LPLV (N = 44)
 - Safety data reported here are from active follow-up (final data cutoff to LPLV; n = 34)

RESULTS

Patients

- 44 patients were re-treated with radium-223 (**Table 1**)
 - Median time from end of initial radium-223 treatment to re-treatment was 6 months (range, 1.2-17.1 mo)
 - All patients had ≥ 2 prior hormonal regimens; 32 of 44 (73%) had failed hormonal agents (ie, abiraterone or enzalutamide)
 - 20 of 44 (45%) had ≥ 1 prior chemotherapy regimen
 - 29 of 44 (66%) completed re-treatment with all 6 injections
- 34 of 44 (77%) patients entered the 2-year active follow-up

Table 1. Demographics and Baseline Characteristics

	Re-treatment N = 44
Age, median (range), y	71 (52-91)
ECOG PS, n (%)	
0	14 (32)
1	27 (61)
2	3 (7)
Extent of disease, bone metastases, n (%)	
< 6	18 (41)
6-20	15 (34)
> 20, not superscan	6 (14)
Superscan	5 (11)
Prior systemic anticancer therapies, n (%)	
Docetaxel	20 (45)
Abiraterone	27 (61)
Enzalutamide	13 (30)
Prior bone-supportive therapies, n (%)	
Bisphosphonates	5 (11)
Denosumab	21 (48)
Laboratory values, median (range)	
Hemoglobin, g/dL	12 (9-16)
Albumin, g/L	39 (32-44)
PSA, µg/L	68 (< 1-2349)
LDH, U/L	203 (115-532)
tALP, U/L	85 (29-705)

ECOG PS = Eastern Cooperative Oncology Group performance status; LDH = lactate dehydrogenase; PSA = prostate-specific antigen; tALP = total alkaline phosphatase.

Safety

- No new safety concerns were observed during active follow-up
 - One treatment-related AE was reported: anemia in 1 of 34 (3%) patients; this event was a worsening of an existing AE
 - No serious drug-related AEs were reported
- At the end of active follow-up, 2 of 34 (6%) patients had grade 1 anemia; no patients had any grade 1-4 neutropenia or thrombocytopenia
- One new primary malignancy was reported: basal cell carcinoma in 1 of 34 (3%) patients; it was diagnosed 12 days after the first radium-223 dose and was determined by the investigator to be unrelated to the study drug
- 28 of 44 (64%) enrolled patients died; 23 of 28 (82%) deaths occurred during active follow-up; the most common cause of death was progressive disease

Exploratory Efficacy

Figure 3. Kaplan-Meier Analysis of Time to Radiographic Bone Progression (A) and Radiographic Progression-Free Survival (rPFS) (B)

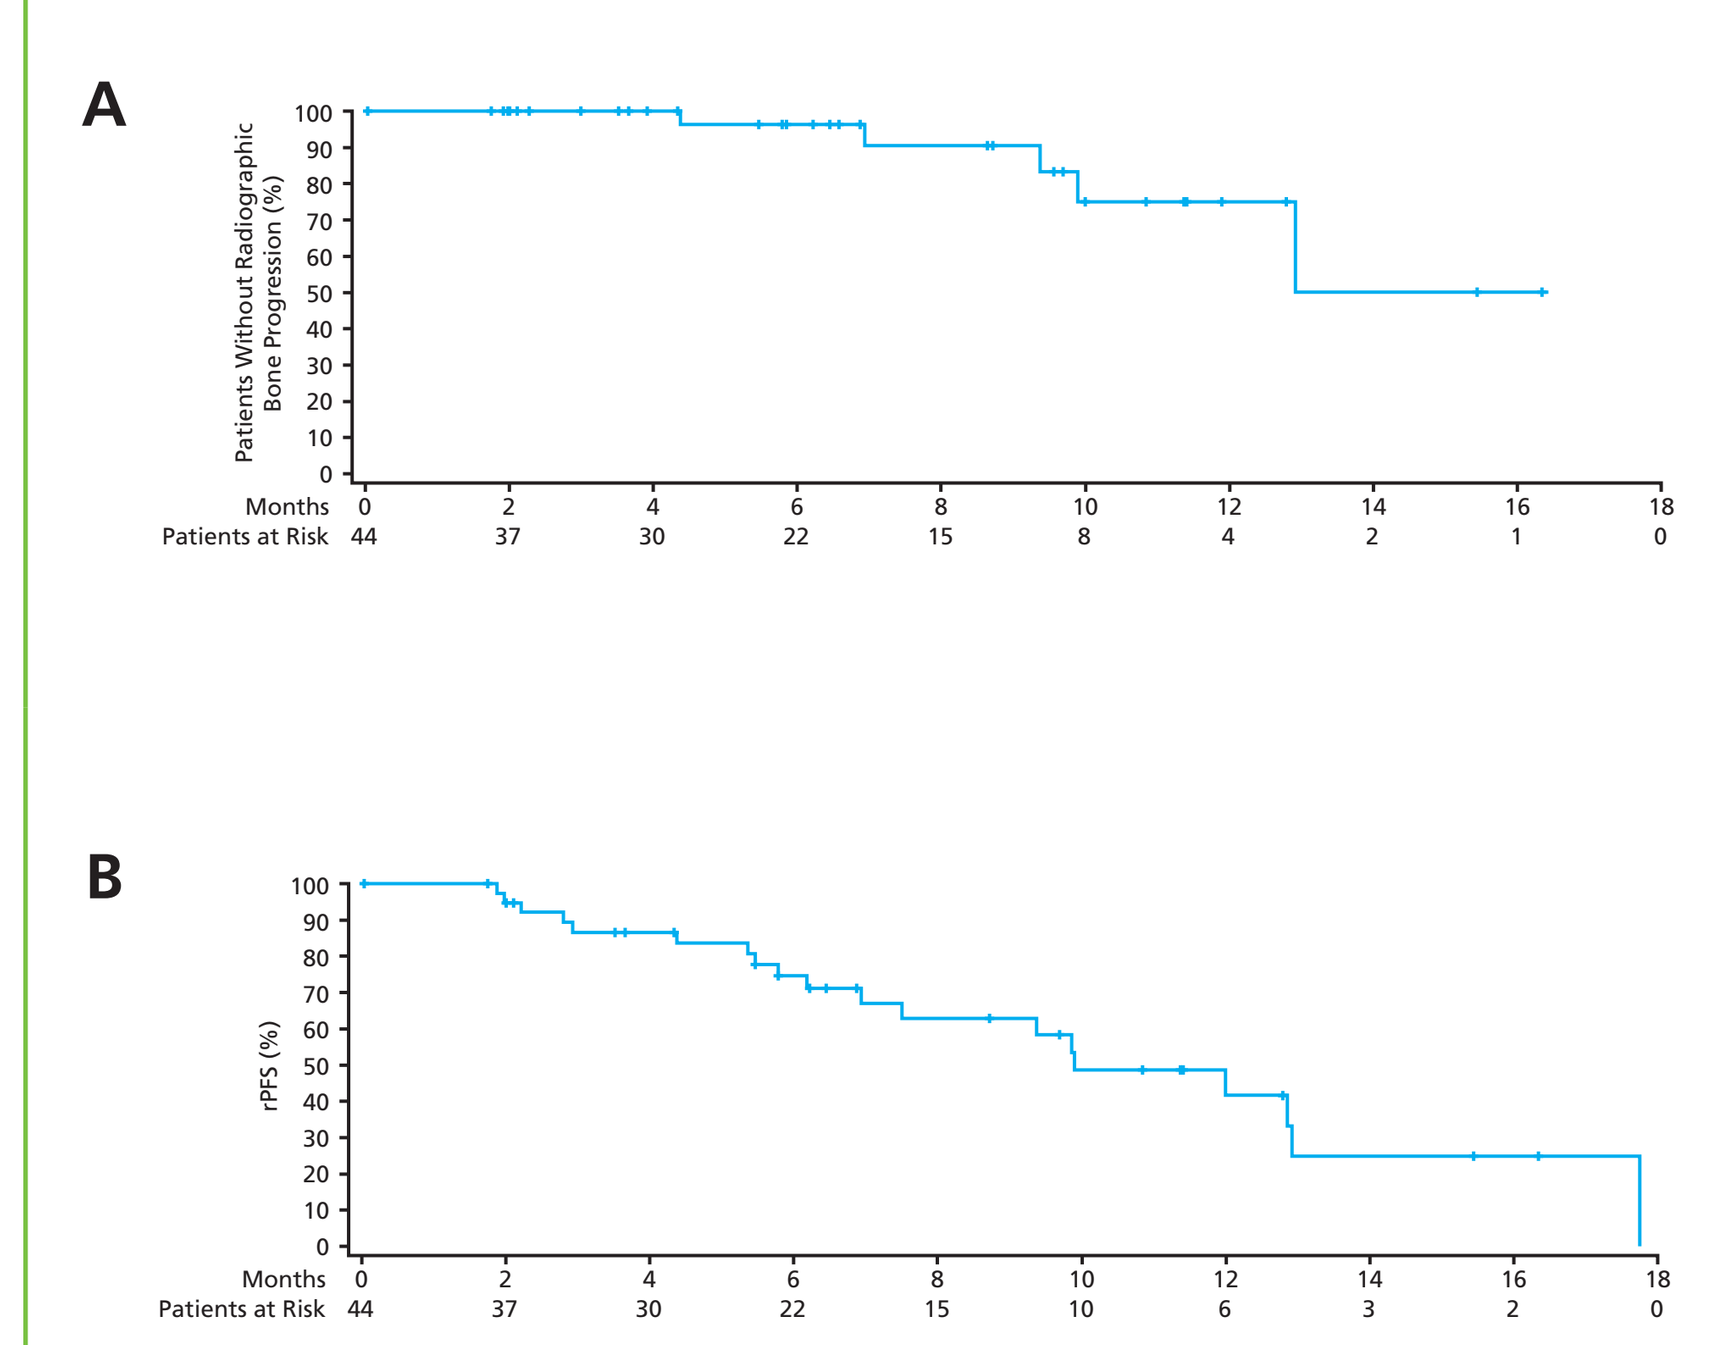
A
Patients Without Radiographic Bone Progression (%)
Months 0 2 4 6 8 10 12 14 16 18
Patients at Risk 44 37 30 22 15 8 4 2 1 0
B
rPFS (%)
Months 0 2 4 6 8 10 12 14 16 18
Patients at Risk 44 37 30 22 15 10 6 3 2 0

- Median time to radiographic bone progression was not reached (**Figure 3A**)
 - Maximum follow-up time for radiographic bone progression was 16.3 months
 - 5 of 44 (11%) patients had radiographic bone progression
- Median radiographic progression-free survival (rPFS) was 9.9 months (**Figure 3B**)
 - Maximum follow-up time for rPFS was 17.7 months
 - 19 of 44 (43%) patients had an rPFS event (radiographic progression or death)

Figure 4. Kaplan-Meier Analysis of Time to Total Alkaline Phosphatase (tALP) (A) and Prostate-Specific Antigen (PSA) (B) Progression

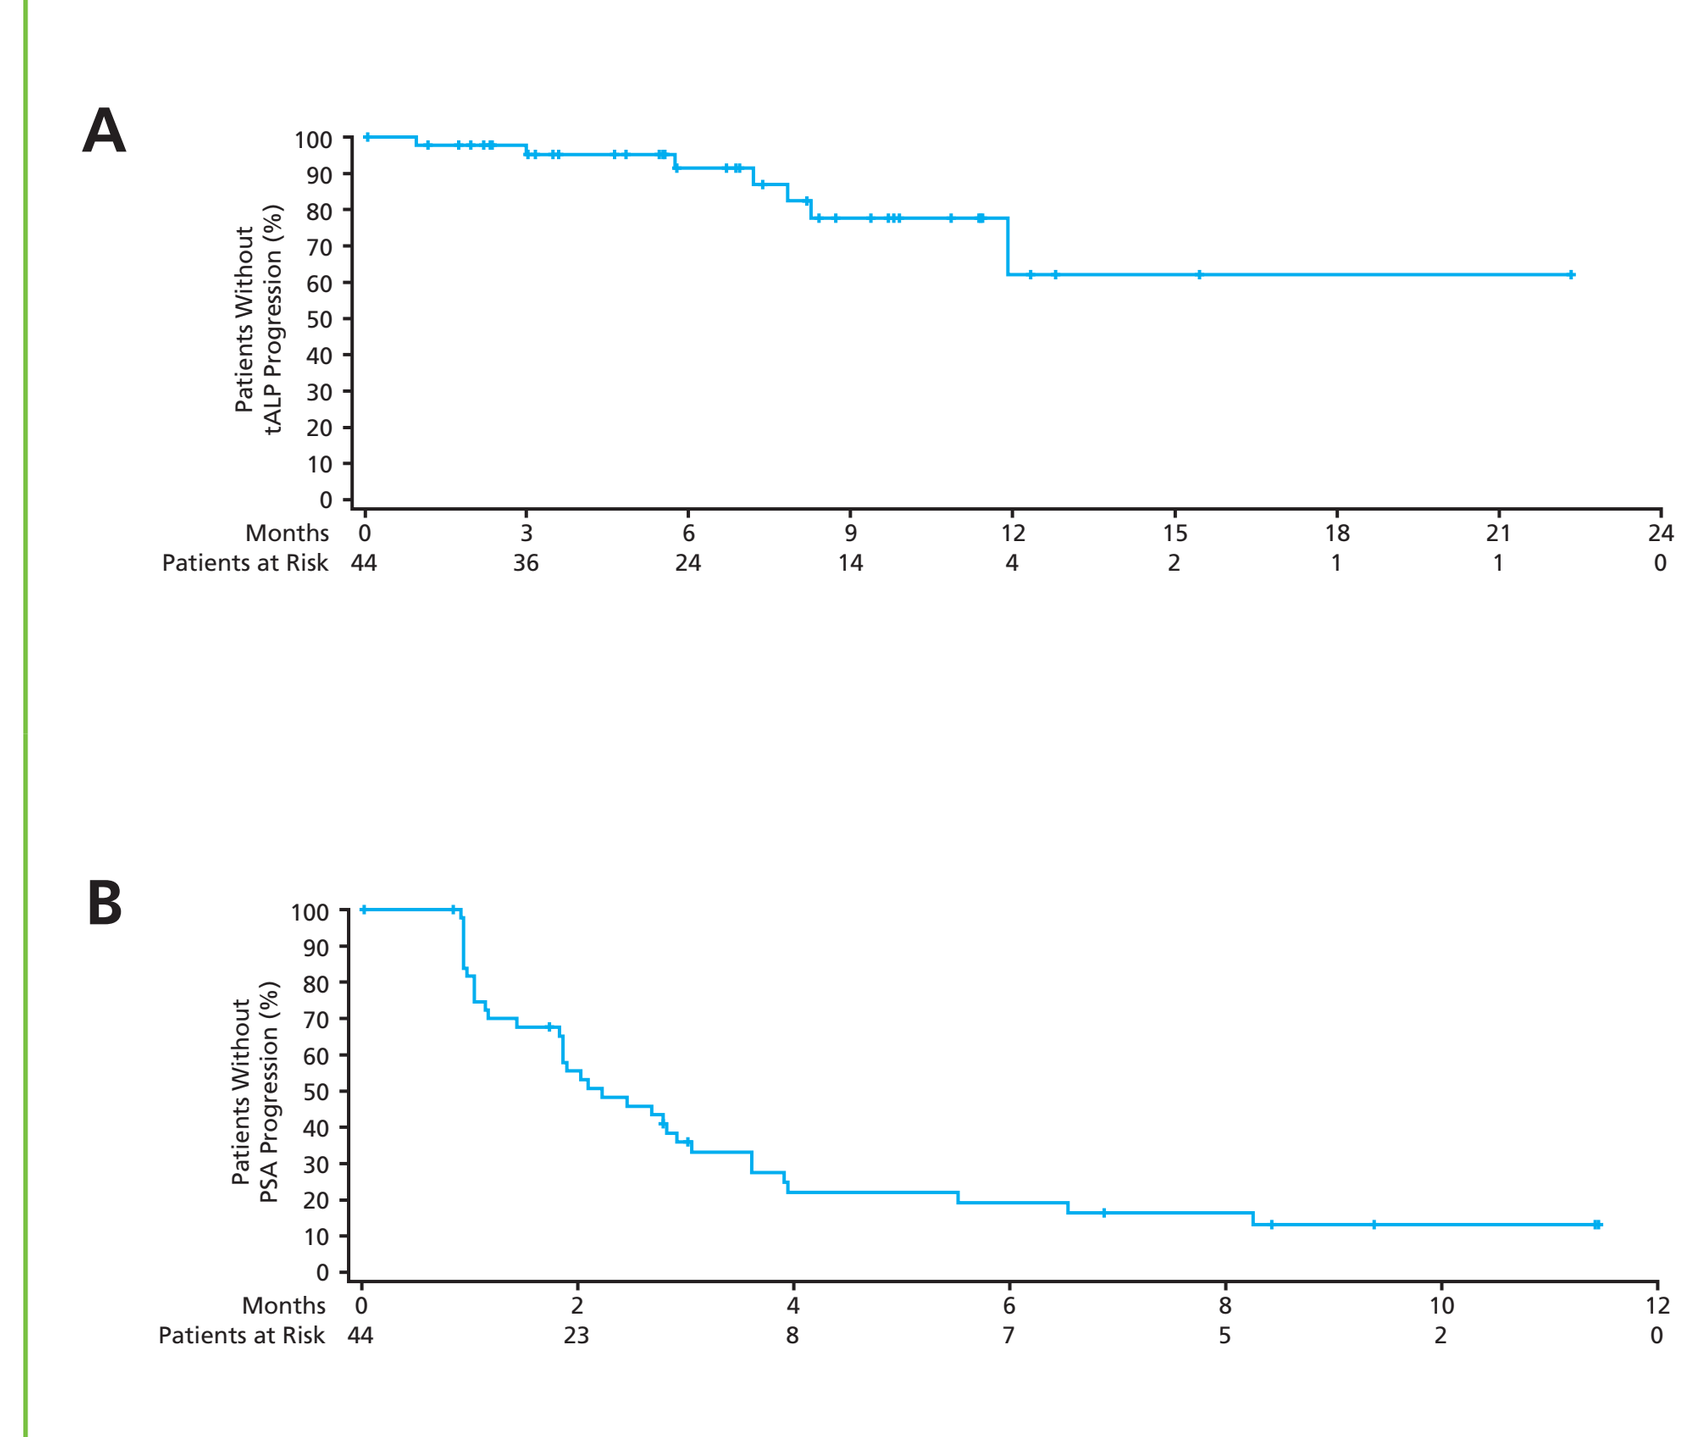
A
Patients Without tALP Progression (%)
Months 0 3 6 9 12 15 18 21 24
Patients at Risk 44 36 24 14 4 2 1 1 0
B
Patients Without PSA Progression (%)
Months 0 2 4 6 8 10 12
Patients at Risk 44 23 8 7 5 2 0

- Median time to tALP progression was not reached by the end of the 2-year follow-up (**Figure 4A**)
 - Maximum follow-up time for tALP progression was 22.3 months
 - 7 of 44 (16%) patients had tALP progression
- Median time to PSA progression was 2.2 months (**Figure 4B**)
 - Maximum follow-up time for PSA progression was 11.4 months
 - 35 of 44 (80%) patients had PSA progression

Figure 5. Kaplan-Meier Analysis of Overall Survival

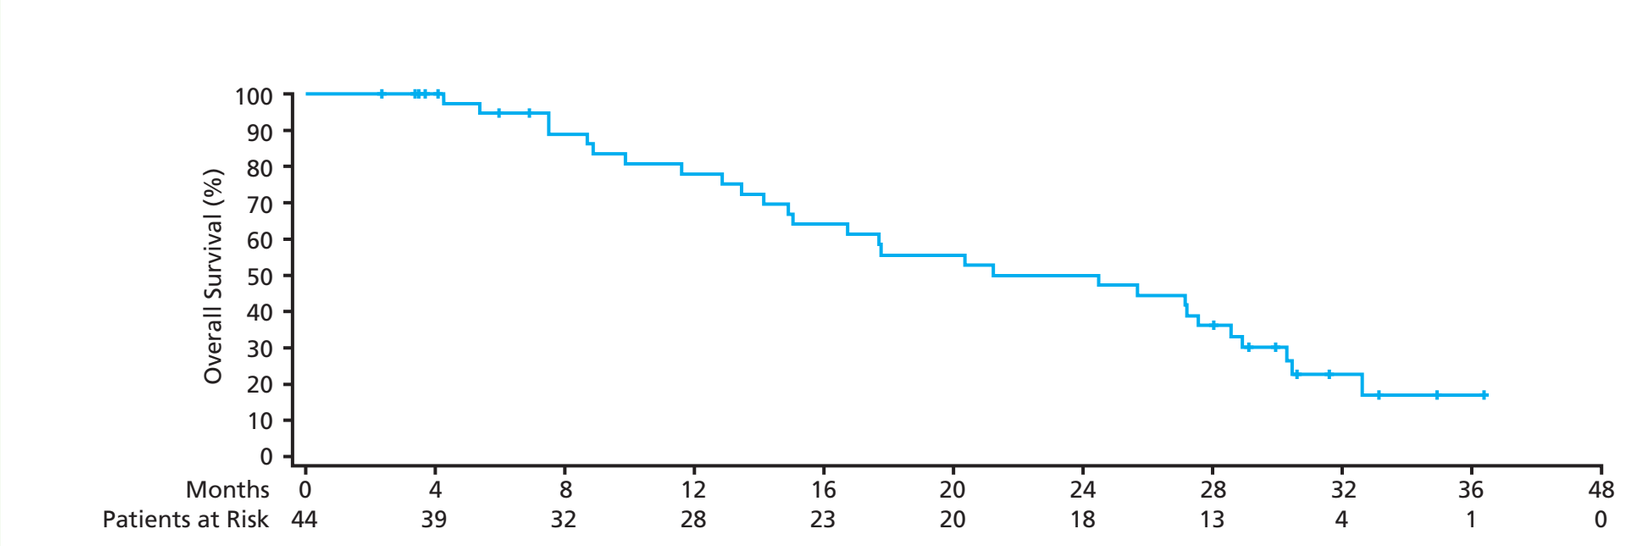
Overall Survival (%)
Months 0 4 8 12 16 20 24 28 32 36 48
Patients at Risk 44 39 32 28 23 20 18 13 4 1 0

- Median overall survival (OS) was 24.4 months (**Figure 5**)
 - Median follow-up for OS was 31.6 months
 - 28 of 44 (64%) patients died

Figure 6. Kaplan-Meier Analysis of Time to First Symptomatic Skeletal Event (SSE) (A) and SSE-Free Survival (SSE-FS) (B)

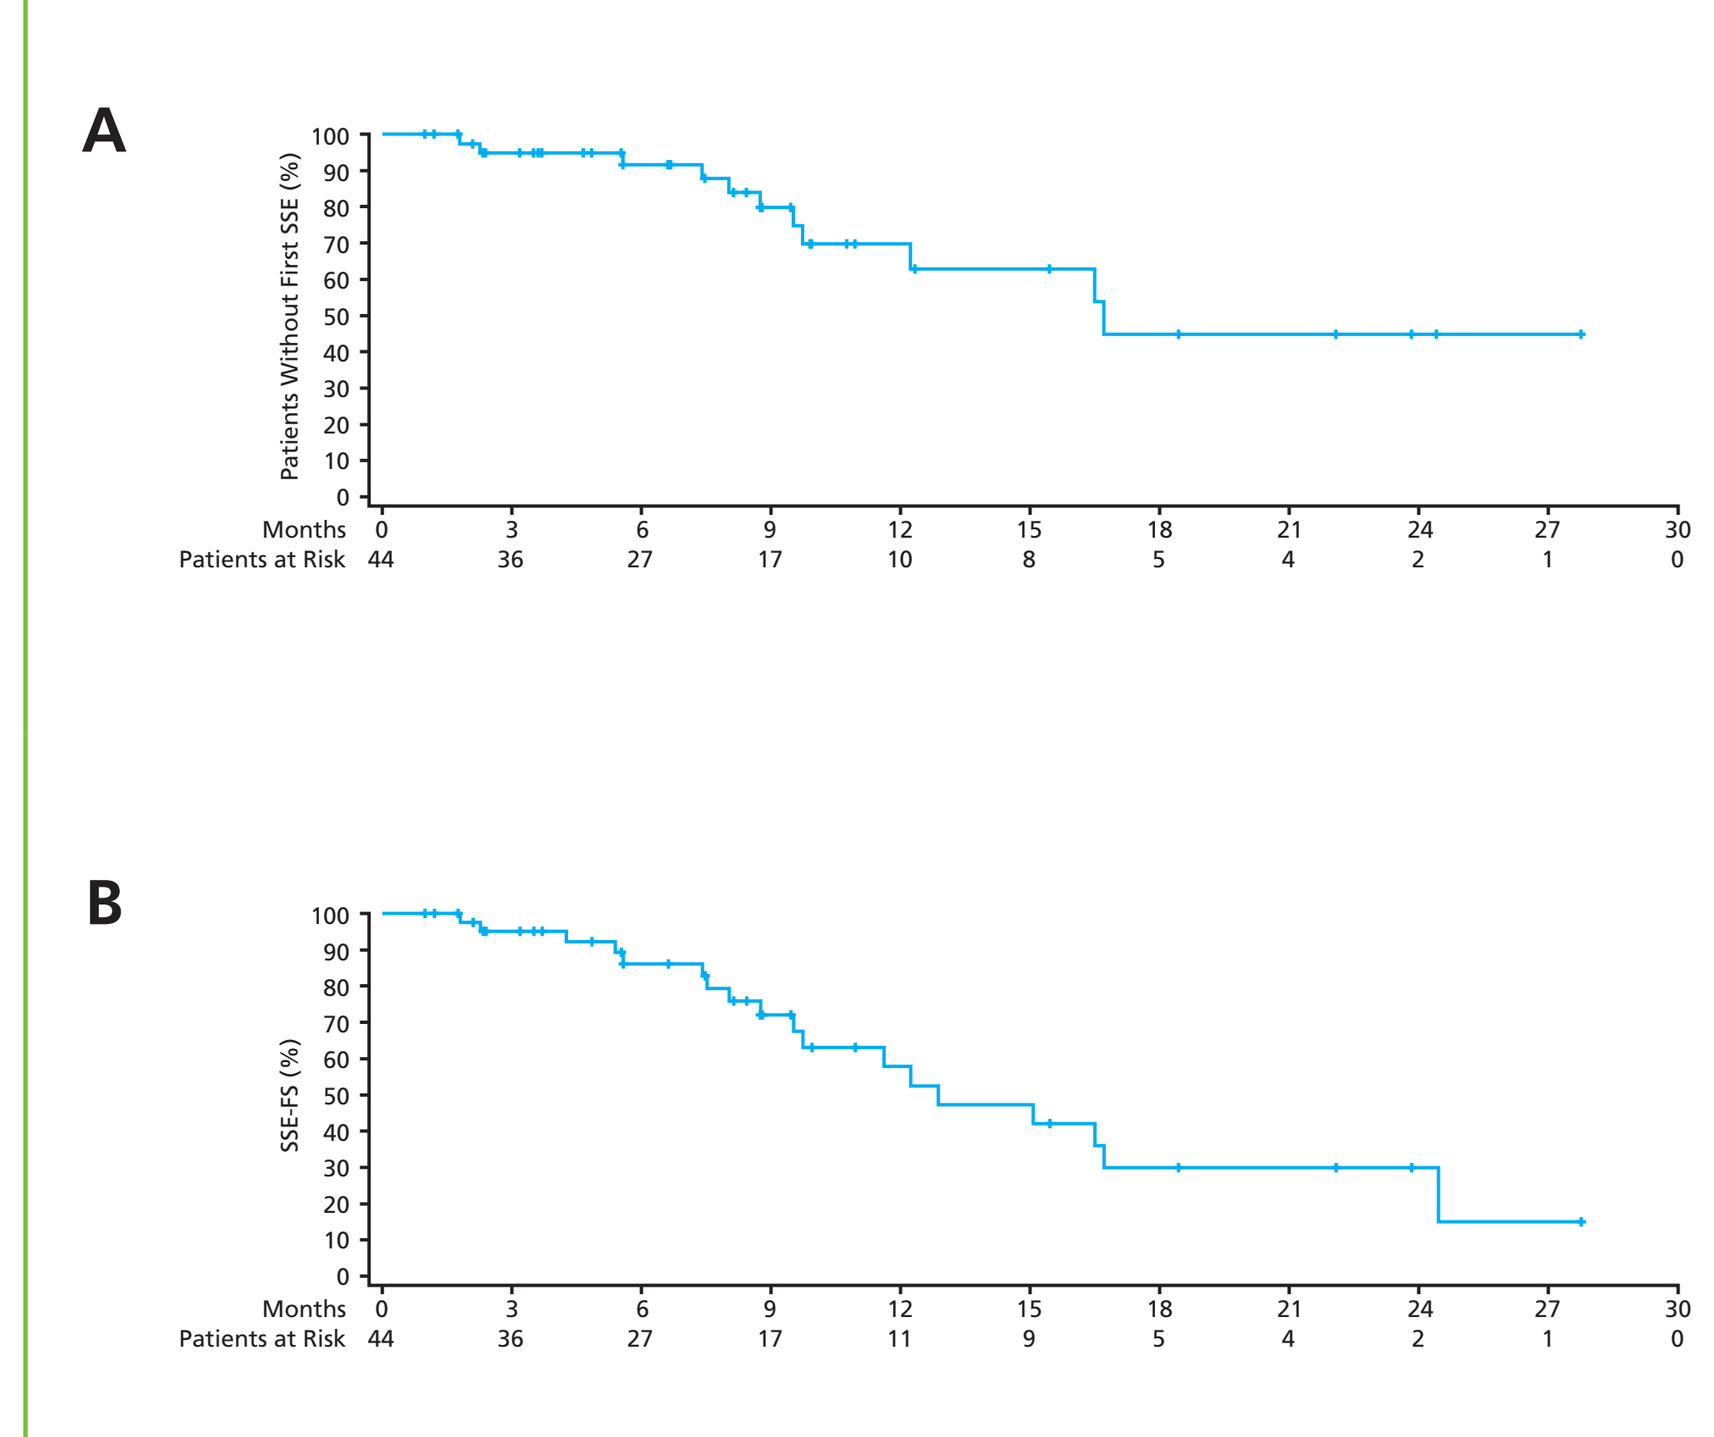
A
Patients Without First SSE (%)
Months 0 3 6 9 12 15 18 21 24 27 30
Patients at Risk 44 36 27 17 10 8 5 4 2 1 0
B
SSE-FS (%)
Months 0 3 6 9 12 15 18 21 24 27 30
Patients at Risk 44 36 27 17 11 9 5 4 2 1 0

- Median time to first symptomatic skeletal event (SSE) was 16.7 months (**Figure 6A**)
 - 14 of 44 (32%) patients experienced at least 1 SSE
 - 3 patients were censored because of new cancer treatment, leaving 11 of 44 (25%) patients for inclusion in the Kaplan-Meier analysis
 - First SSEs were external beam radiation therapy for bone pain (10 of 44 [23%]), pathologic bone fracture (3 of 44 [7%]), and spinal cord compression (1 of 44 [2%])
- Median SSE-free survival (SSE-FS) was 12.8 months (**Figure 6B**)
 - 18 of 44 (41%) patients had an SSE-FS event (SSE or death)
- Maximum follow-up time for SSEs and SSE-FS was 27.8 months

CONCLUSIONS

- Re-treatment with radium-223 was well tolerated in this selected patient population and led to minimal hematologic toxicity
- OS and SSE-FS results indicate that re-treatment with radium-223 may be an option for CRPC patients with bone metastases who previously benefited from a first radium-223 treatment course; this requires further evaluation in larger prospective trials

Trial (NCT01934790) sponsored by Bayer HealthCare Pharmaceuticals.
The authors wish to thank SciStrategy Communications for assistance in the development of this poster.
For questions or information regarding this poster, please contact Dr Neil Mariados at nmariados@ampofny.com.

Poster presented at 2018 Annual Meeting of the American Urological Association (AUA), San Francisco, California.
Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.