MP42-08

Renal Cancer Surgery for Patients without Preexisting Chronic Kidney Disease: Is there a Survival Benefit for Elective Partial Nephrectomy?

Cleveland Clinic

Suk-Ouichai C, Tanaka H, Wu J, Caraballo E, Demirjian S, Li J, Isharwal S, Campbell SC

Department of Urology, Glickman Urological and Kidney Institute, Cleveland Clinic, Cleveland, OH

BACKGROUND

- Retrospective studies suggest that partial nephrectomy (PN) provides improved survival compared to radical nephrectomy (RN), even when performed electively
- Selection bias may be a contributing factor and further investigation is required
- We evaluated factors associated with non-renal cancer-related mortality (NRCM) after elective PN or RN for patients with **preoperative GFR ≥60ml/min/1.73m²**

METHODS

- Patients with **preoperative GFR ≥60ml/min/1.73m²** managed with elective PN or RN for presumed renal cancer at our center from 1997-2008 were retrospective review
- **Non-renal cancer-related mortality (NRCM)** was chosen for the primary endpoint because it removes any potential oncological imbalances between the various cohorts and more accurately reflects the impact of renal function and its metabolic consequences
- NRCM was analyzed by Kaplan-Meier in various cohorts based on functional parameters including preoperative and new baseline GFR but also by procedure (PN versus RN)
- Cox proportional hazards assessed factors associated with NRCM among patients with **new baseline GFR≥45ml/min/1.73m²**

RESULTS

- Of 3,133 patients, PN was performed in 1,732 cases (55%) and RN in 1,401 cases (45%). Median age was 59 years and 60% of patients were male. (**Table 1**)
- Median preoperative GFR was 85 ml/min/1.73m² in both the PN and RN cohorts
- New baseline GFR after surgery was 80 ml/min/1.73m² for PN and 63 ml/min/1.73m² for RN (p<0.001)
- Median follow-up was 9.3 years
- Patients with preoperative GFR less than 75ml/min/1.73m² had significantly higher NRSM (p=0.001)(**Figure 1A**)
- NRCM at 10-years was 11.4% after PN and 19.0% after RN (p<0.001)(**Figure 1B**)
- NRCM at 10 years was similar for all cohorts of patients with new baseline GFR ≥45 ml/min/1.73m² (11.3-16.3%, p=0.26), but fell substantially for patients with new baseline GFR below this level (25.3%, p=0.001)(**Figure 1C**)
- Excluding patients with new baseline GFR <45ml/min/1.73m² (n=290), NRCM at 10-years was still substantially improved after PN than RN (10.7% vs. 17.6%, p<0.001)(**Figure 2B**)
- For patients with new baseline GFR ≥45ml/min/1.73m², age, gender, and procedure (PN versus RN) were associated with NRCM (all p≤0.001) on univariable and multivariable analyses (**Tables 2 and 3**)
- In contrast, on multivariable analysis, increased new baseline GFR, as would be seen with PN, failed to associate with improved NRCM

RESULTS

Figure 1. Analysis of all patients without preoperative CKD (n=3133)

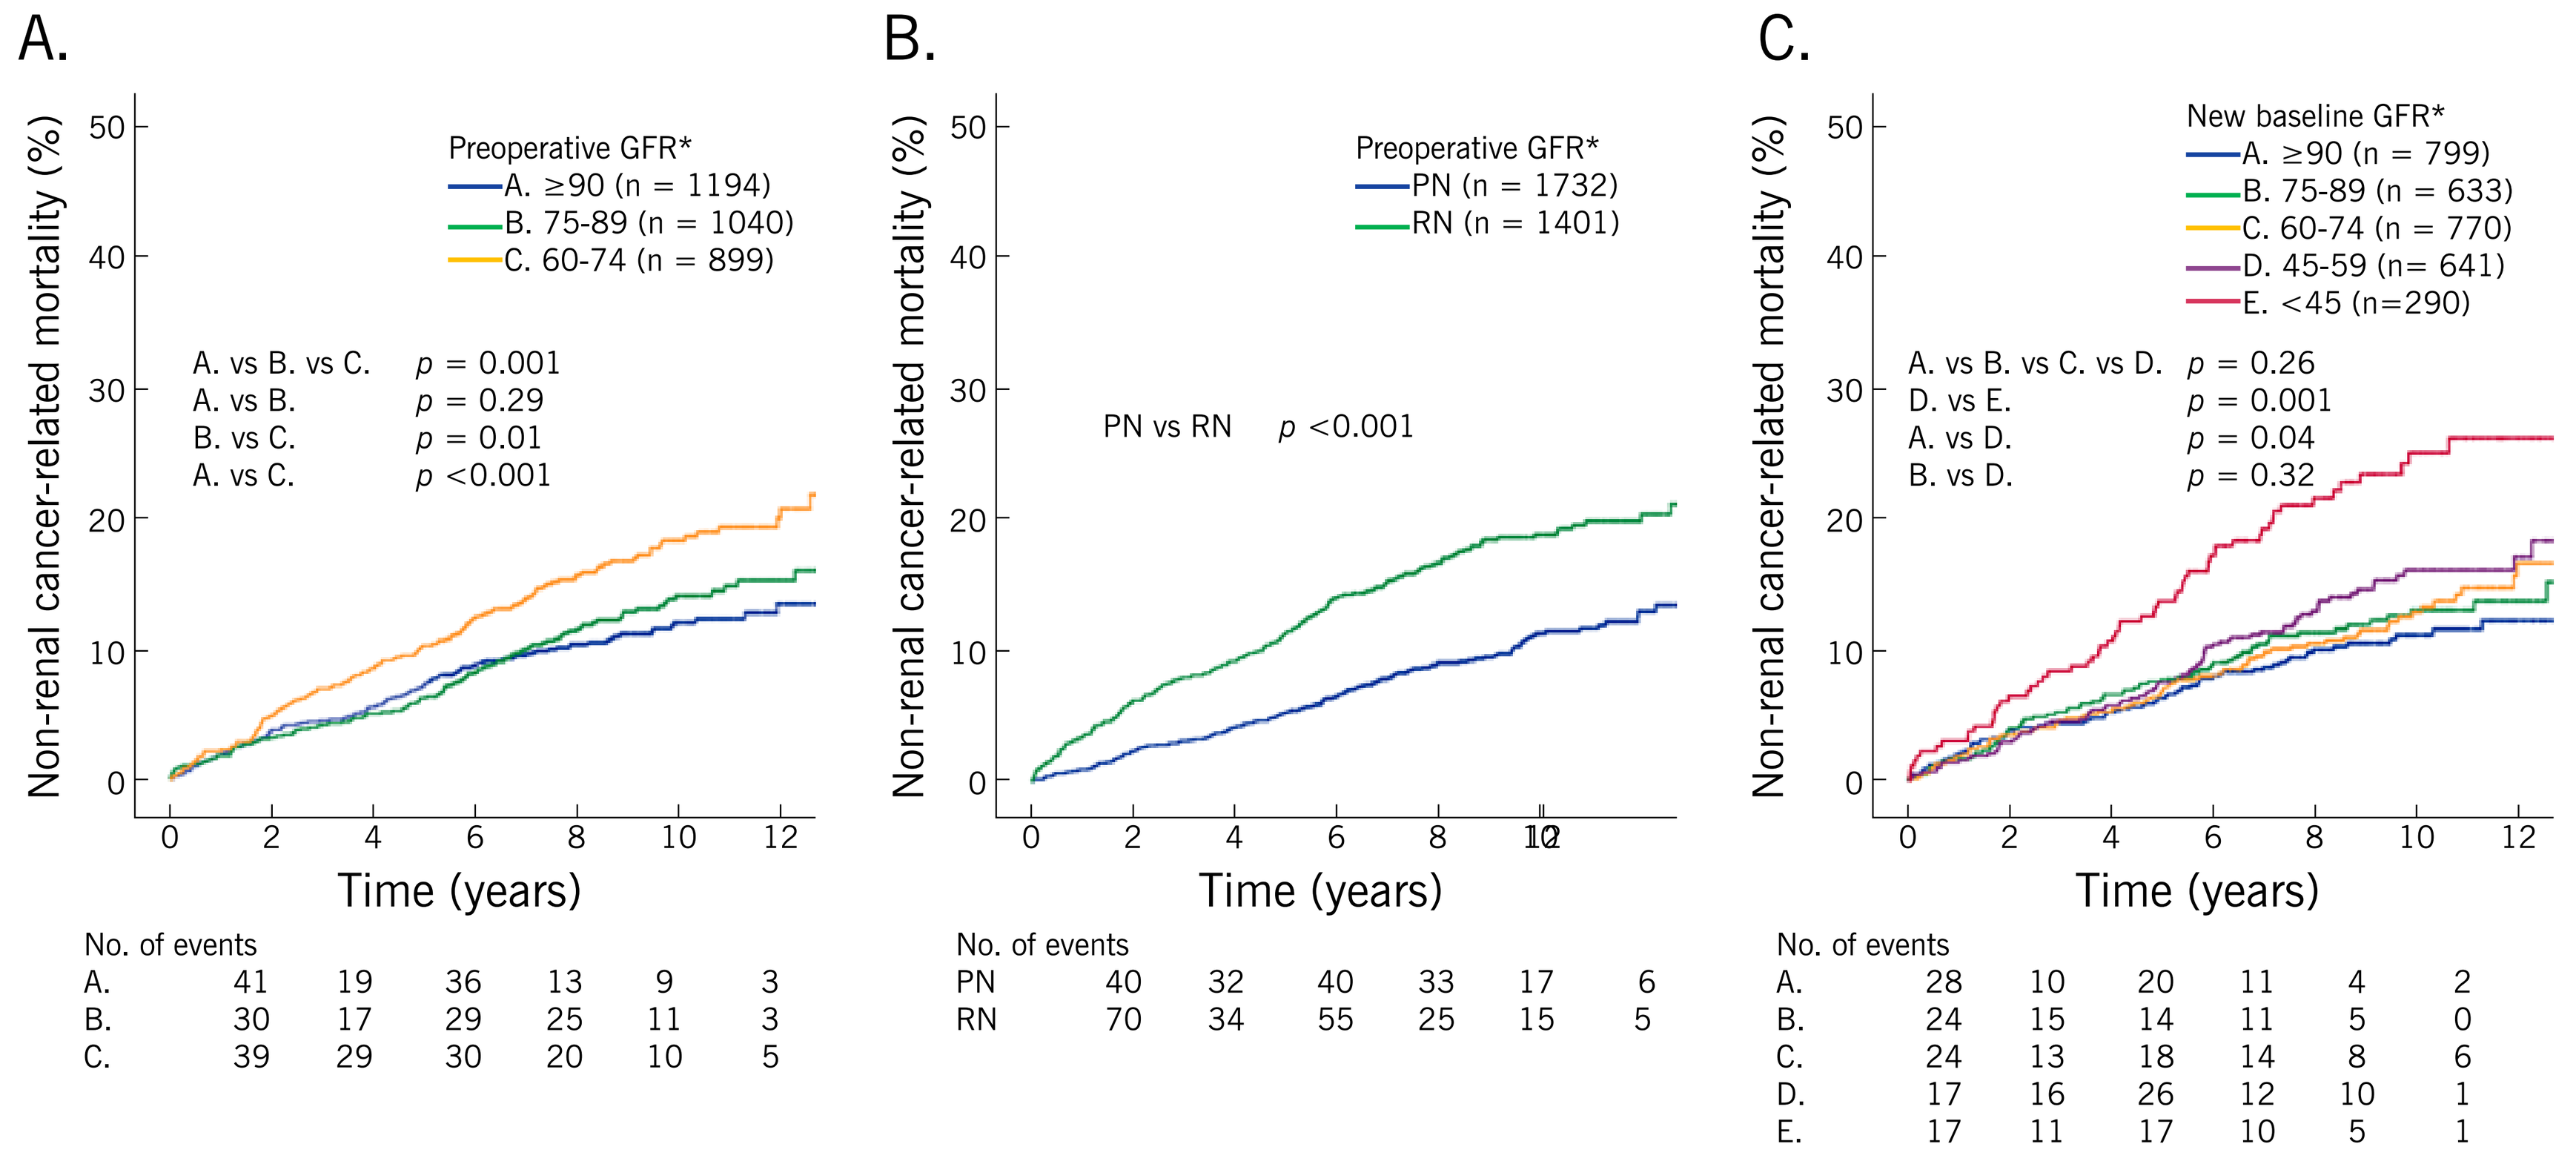

A.
Non-renal cancer-related mortality (%)
Preoperative GFR*
A. ≥90 (n = 1194)
B. 75-89 (n = 1040)
C. 60-74 (n = 899)
A. vs B. vs C. p = 0.001
A. vs B. p = 0.29
B. vs C. p = 0.01
A. vs C. p <0.001
Time (years)
No. of events
A. 41 19 36 13 9 3
B. 30 17 29 25 11 3
C. 39 29 30 20 10 5
B.
Non-renal cancer-related mortality (%)
Preoperative GFR*
PN (n = 1732)
RN (n = 1401)
PN vs RN p <0.001
Time (years)
No. of events
PN 40 32 40 33 17 6
RN 70 34 55 25 15 5
C.
Non-renal cancer-related mortality (%)
New baseline GFR*
A. ≥90 (n = 799)
B. 75-89 (n = 633)
C. 60-74 (n = 770)
D. 45-59 (n= 641)
E. <45 (n=290)
A. vs B. vs C. vs D. p = 0.26
D. vs E. p = 0.001
A. vs D. p = 0.04
B. vs D. p = 0.32
Time (years)
No. of events
A. 28 10 20 11 4 2
B. 24 15 14 11 5 0
C. 24 13 18 14 8 6
D. 17 16 26 12 10 1
E. 17 11 17 10 5 1

Table 1. Patient Characteristics

Overall		Renal cancer surgery PN	RN	*p* value
Patients (%)	3133	1732 (55)	1401 (45)	
Age (years) (median, IQR)	59 (50-67)	58 (49-66)	60 (50-68)	<0.001
Male (%)	1894 (60)	1058 (61)	836 (60)	0.53
African-American (%)	204 (7)	103 (6)	101 (7)	0.16
Hypertension (%)	1097 (35)	692 (40)	405 (29)	<0.001
Diabetes (%)	355 (11)	224 (13)	131 (9)	0.002
Heart disease (%)	32 (1)	16 (1)	16 (2)	0.54
Preoperative GFR* (median, IQR)	85 (73-97)	85 (73-98)	85 (72-96)	0.11
Preoperative GFR (%)				0.028
≥ 90 ml/min/1.73m²		1194 (38)	696 (40)	498 (36)
75-89 ml/min/1.73m²		1040(33)	552 (32)	488 (35)
60-74 ml/min/1.73m²		899(29)	484 (28)	415 (29)
New baseline GFR* (median, IQR)	72 (57-90)	80 (64-96)	63 (51-79)	<0.001
New baseline GFR (%)				<0.001
≥ 90 ml/min/1.73m²	799 (26)	588 (34)	211 (15)	
75-89 ml/min/1.73m²	633 (20)	437 (25)	196 (14)	
60-74 ml/min/1.73m²	770 (25)	372 (22)	398 (28)	
45-59 ml/min/1.73m²	641 (20)	226 (13)	415 (30)	
< 45 ml/min/1.73m²	290 (9)	109 (6)	181 (13)	
% Global GFR preserved (median, IQR)	87 (70-100)	95 (80-105)	74 (63-90)	<0.001
Follow-up (years) (median, IQR)	9.3 (7.5-11.2)	9.5 (7.8-11.6)	9.0 (6.4-11.2)	
50% decrease in GFR or Dialysis (%)	127 (4)	44 (3)	83 (6)	<0.001
% 10-year overall mortality	23.4	13.7	35.2	<0.001
% 10-year renal cancer specific mortality	10.9	2.7	21.3	<0.001
% 10-year non-renal cancer-related mortality	14.5	11.4	19.0	<0.001

* ml/min/1.73m²

Figure 2. Analysis of patients without preoperative CKD, excluding patients with new baseline GFR <45ml/min/1.73m² (n=2843)

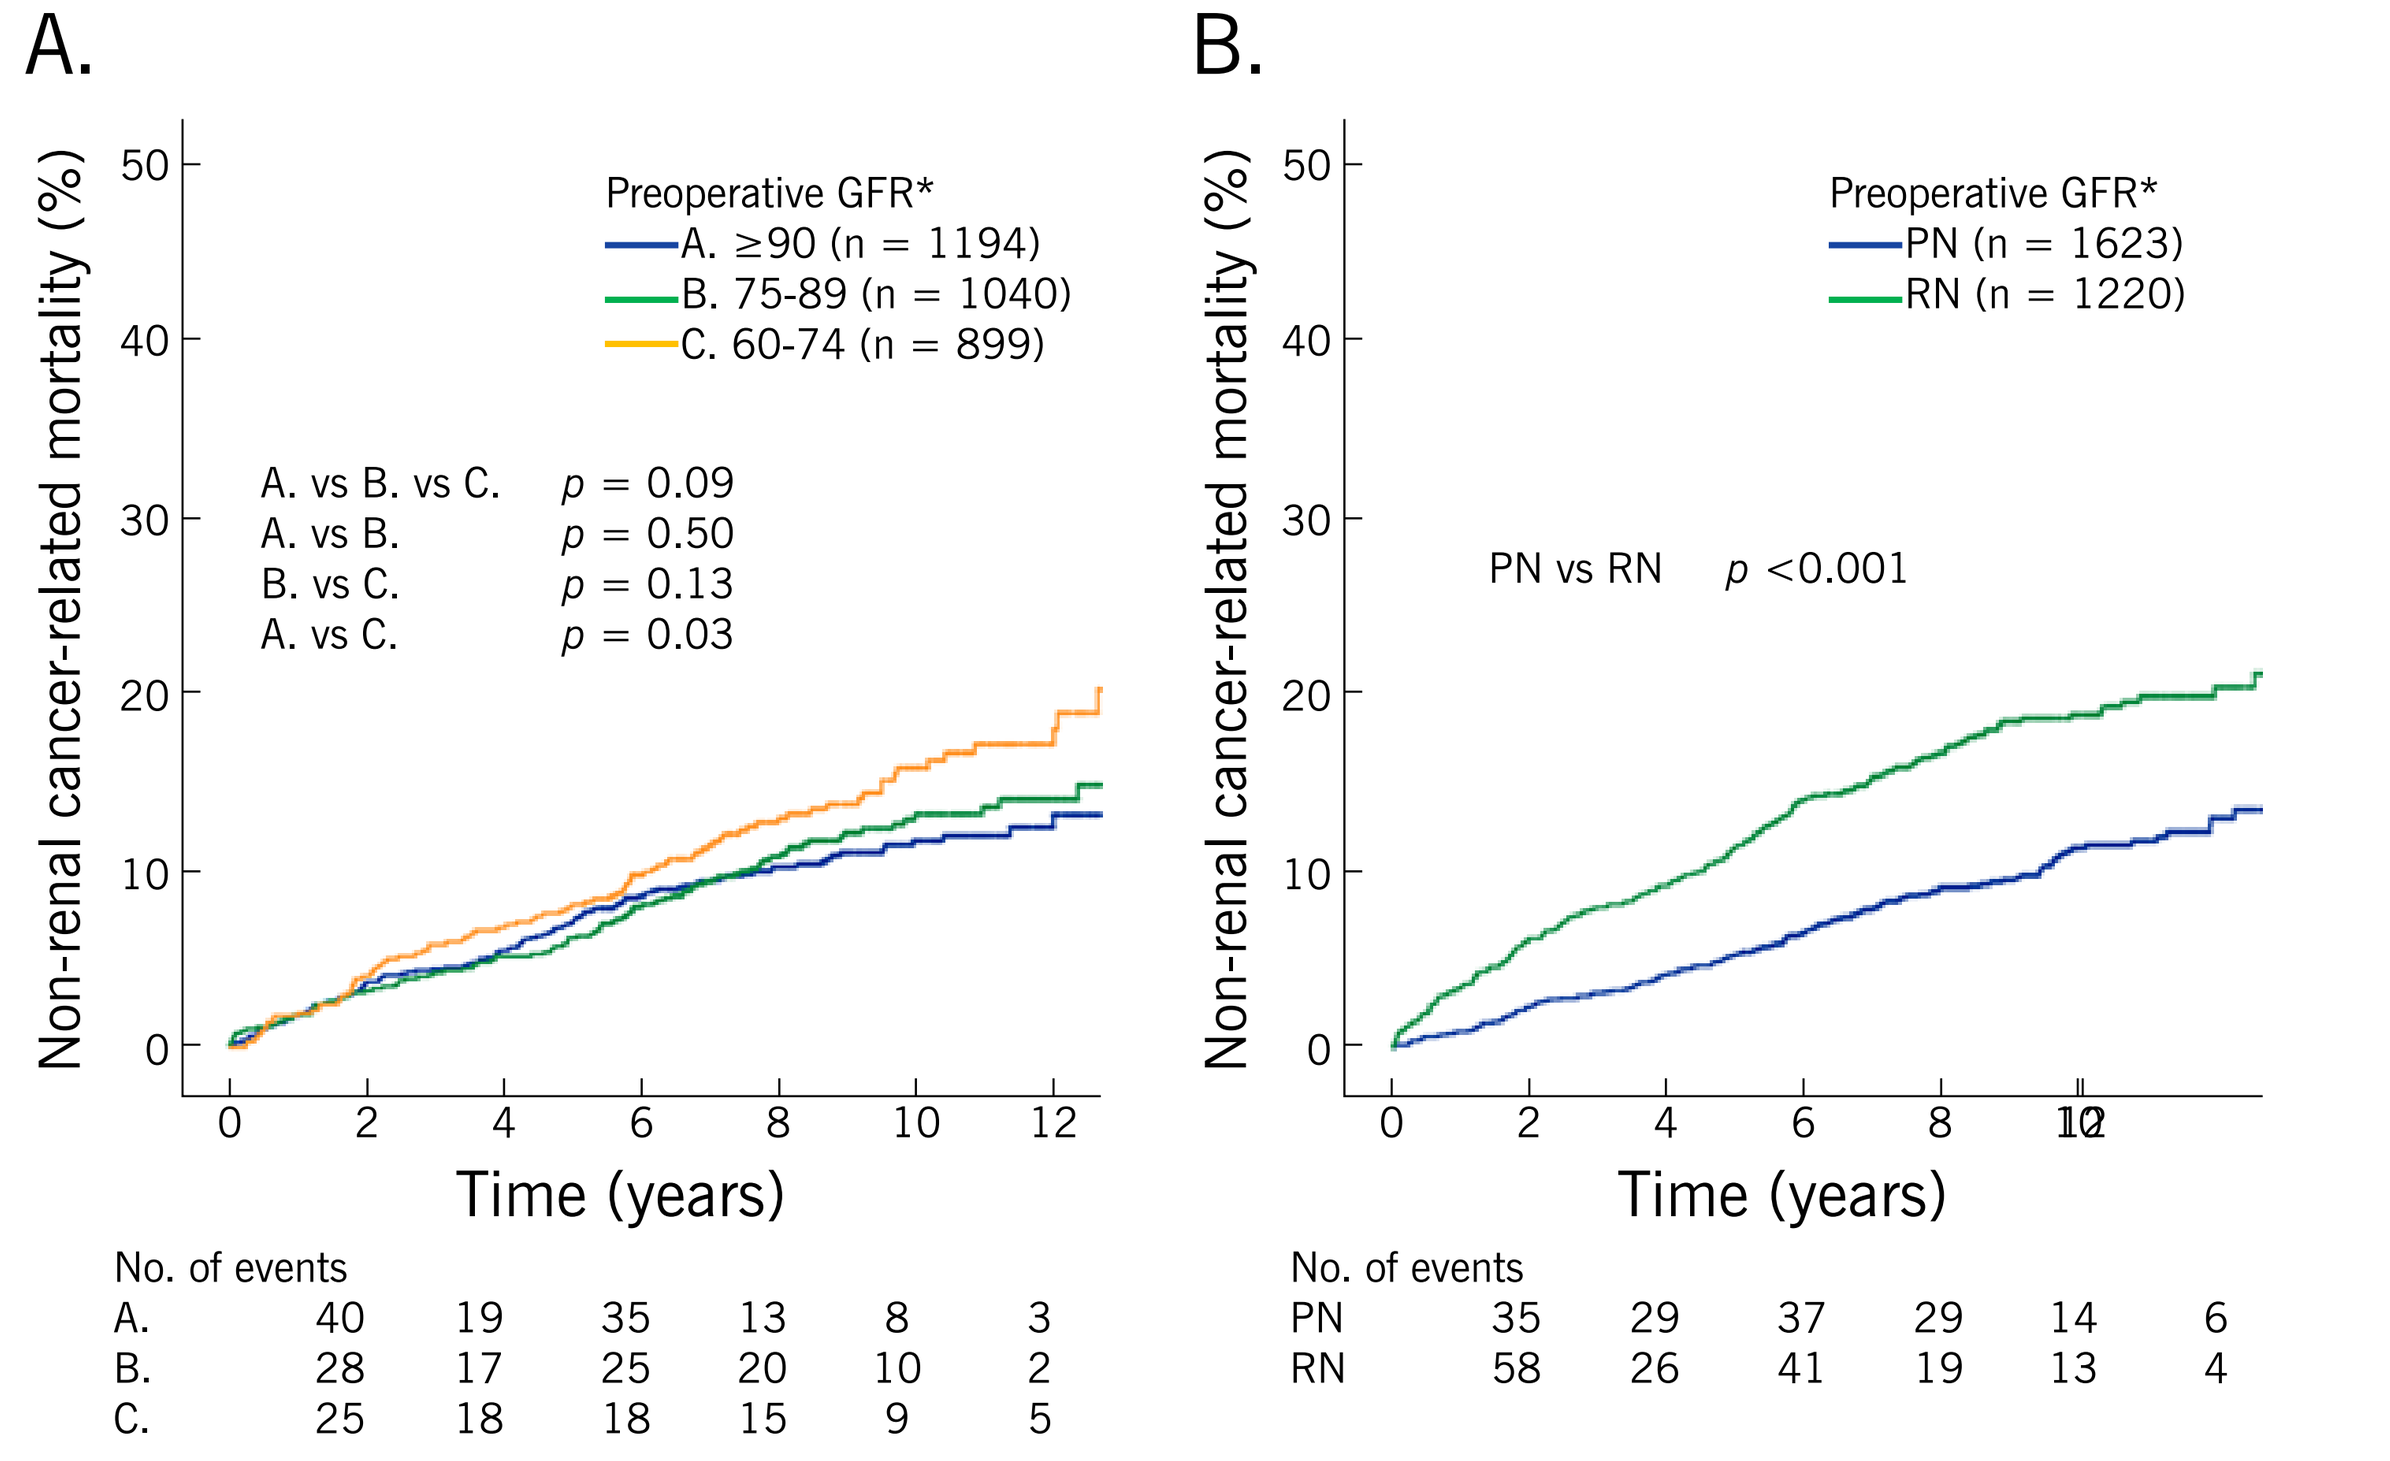

A.
Non-renal cancer-related mortality (%)
Preoperative GFR*
A. ≥90 (n = 1194)
B. 75-89 (n = 1040)
C. 60-74 (n = 899)
A. vs B. vs C. p = 0.09
A. vs B. p = 0.50
B. vs C. p = 0.13
A. vs C. p = 0.03
Time (years)
No. of events
A. 40 19 35 13 8 3
B. 28 17 25 20 10 2
C. 25 18 18 15 9 5
B.
Non-renal cancer-related mortality (%)
Preoperative GFR*
PN (n = 1623)
RN (n = 1220)
PN vs RN p <0.001
Time (years)
No. of events
PN 35 29 37 29 14 6
RN 58 26 41 19 13 4

Table 2. Univariable analysis of NRCM for patients with preoperative GFR ≥60 ml/min/1.73m² and new baseline GFR ≥45 ml/min/1.73m²

Variable	HR	95% CI	*p* value
Age > median of 58 years	2.46	1.94, 3.12	**<0.001**
Male	1.58	1.24, 2.00	**<0.001**
African American	1.23	0.83, 1.83	0.30
Hypertension	1.15	0.92, 1.45	0.23
Diabetes	1.28	0.93, 1.76	0.14
Heart disease	1.98	0.82, 4.79	0.13
Preoperative GFR	0.99	0.98, 1.01	0.11
Radical nephrectomy	1.81	1.45, 2.26	**<0.001**
New baseline GFR	1.00	0.99, 1.01	0.08

Table 3. Multivariable analysis of NRCM for patients with preoperative GFR ≥60 ml/min/1.73m² and new baseline GFR ≥45 ml/min/1.73m²

Inclusion of surgical approach			
Variable	HR	95% CI	*p* value
Age > median of 58 years	2.68	1.92, 3.23	**<0.001**
Male	1.56	1.22, 1.98	**<0.001**
African American	1.16	0.78, 1.74	0.47
Hypertension	0.96	0.75, 1.24	0.79
Diabetes	1.15	0.79, 1.57	0.54
Heart disease	1.77	0.66, 4.10	0.28
Preoperative GFR	1.01	0.99, 1.01	0.15
Radical nephrectomy	1.80	1.44, 2.26	**<0.001**
Inclusion of new baseline GFR			
Variable	HR	95% CI	*p* value
Age > median of 58 years	2.62	2.02, 3.41	**<0.001**
Male	1.53	1.20, 1.95	**0.001**
African American	1.20	0.80, 1.79	0.39
Hypertension	0.90	0.70, 1.15	0.40
Diabetes	1.08	0.76, 1.52	0.67
Heart disease	1.65	0.67, 4.11	0.28
Preoperative GFR	1.01	0.99, 1.01	0.21
New baseline GFR	1.00	0.99, 1.01	0.60

CONCLUSIONS

- For patients with preoperative GFR ≥ 60ml/min/1.73m² undergoing renal cancer surgery, our data suggest priority for achieving new baseline GFR ≥45ml/min/1.73m²
- For this cohort, PN associated with improved survival even though increased new baseline GFR failed to correlate
- Given that the functional dividends of elective PN did not correlate with better survival, our study suggests that selection bias for procedure is the main factor impacting outcomes