18-5523

MP32-10

GLANS PENIS SENSITIVITY TO VIBRATION AFTER INFLATABLE PENILE PROSTHESIS IMPLANTATION USING A PENOSCROTAL AND INFRAPUBIC ACCESS

Nika Akhvlediani, Dmitry Enikeev, Denis Butnaru, Igor Matukhov

SECHENOV UNIVERSITY
Research Institute for Uronephrology and Reproductive Health

INTRODUCTION AND OBJECTIVES:

Three-piece inflatable penile prostheses (IPP) are usually placed via a penoscrotal or infrapubic approach. The latter carries the risk of damage to dorsal nerve fibers potentially leading to a decrease in glans penis sensitivity. The aim of the study was to compare penile sensitivity to vibration (PSV) after IPP implantation via a penoscrotal and infrapubic approach.

METHODS:

This prospective randomized cohort study included 65 patients selected for IPP implantation. The patients were divided into two groups. Group 1 (n=31, aged 47.7±6.2 years) underwent surgery using a penoscrotal approach. Group 2 (n=34, aged 48.1±7.0 years)underwent surgery using a infrapubic approach. Prior to corporotomy, the surgery using a infrapubic approach was performed with the help of 4,5-fold optical magnification to avoid damaging dorsal nerve fibers.

Biothesiometry at 64 Hz was used to assess PSV (dB) prior to and 1.5 months after surgery (when return to normal sexual activity was recommended).

STUDY DESIGN

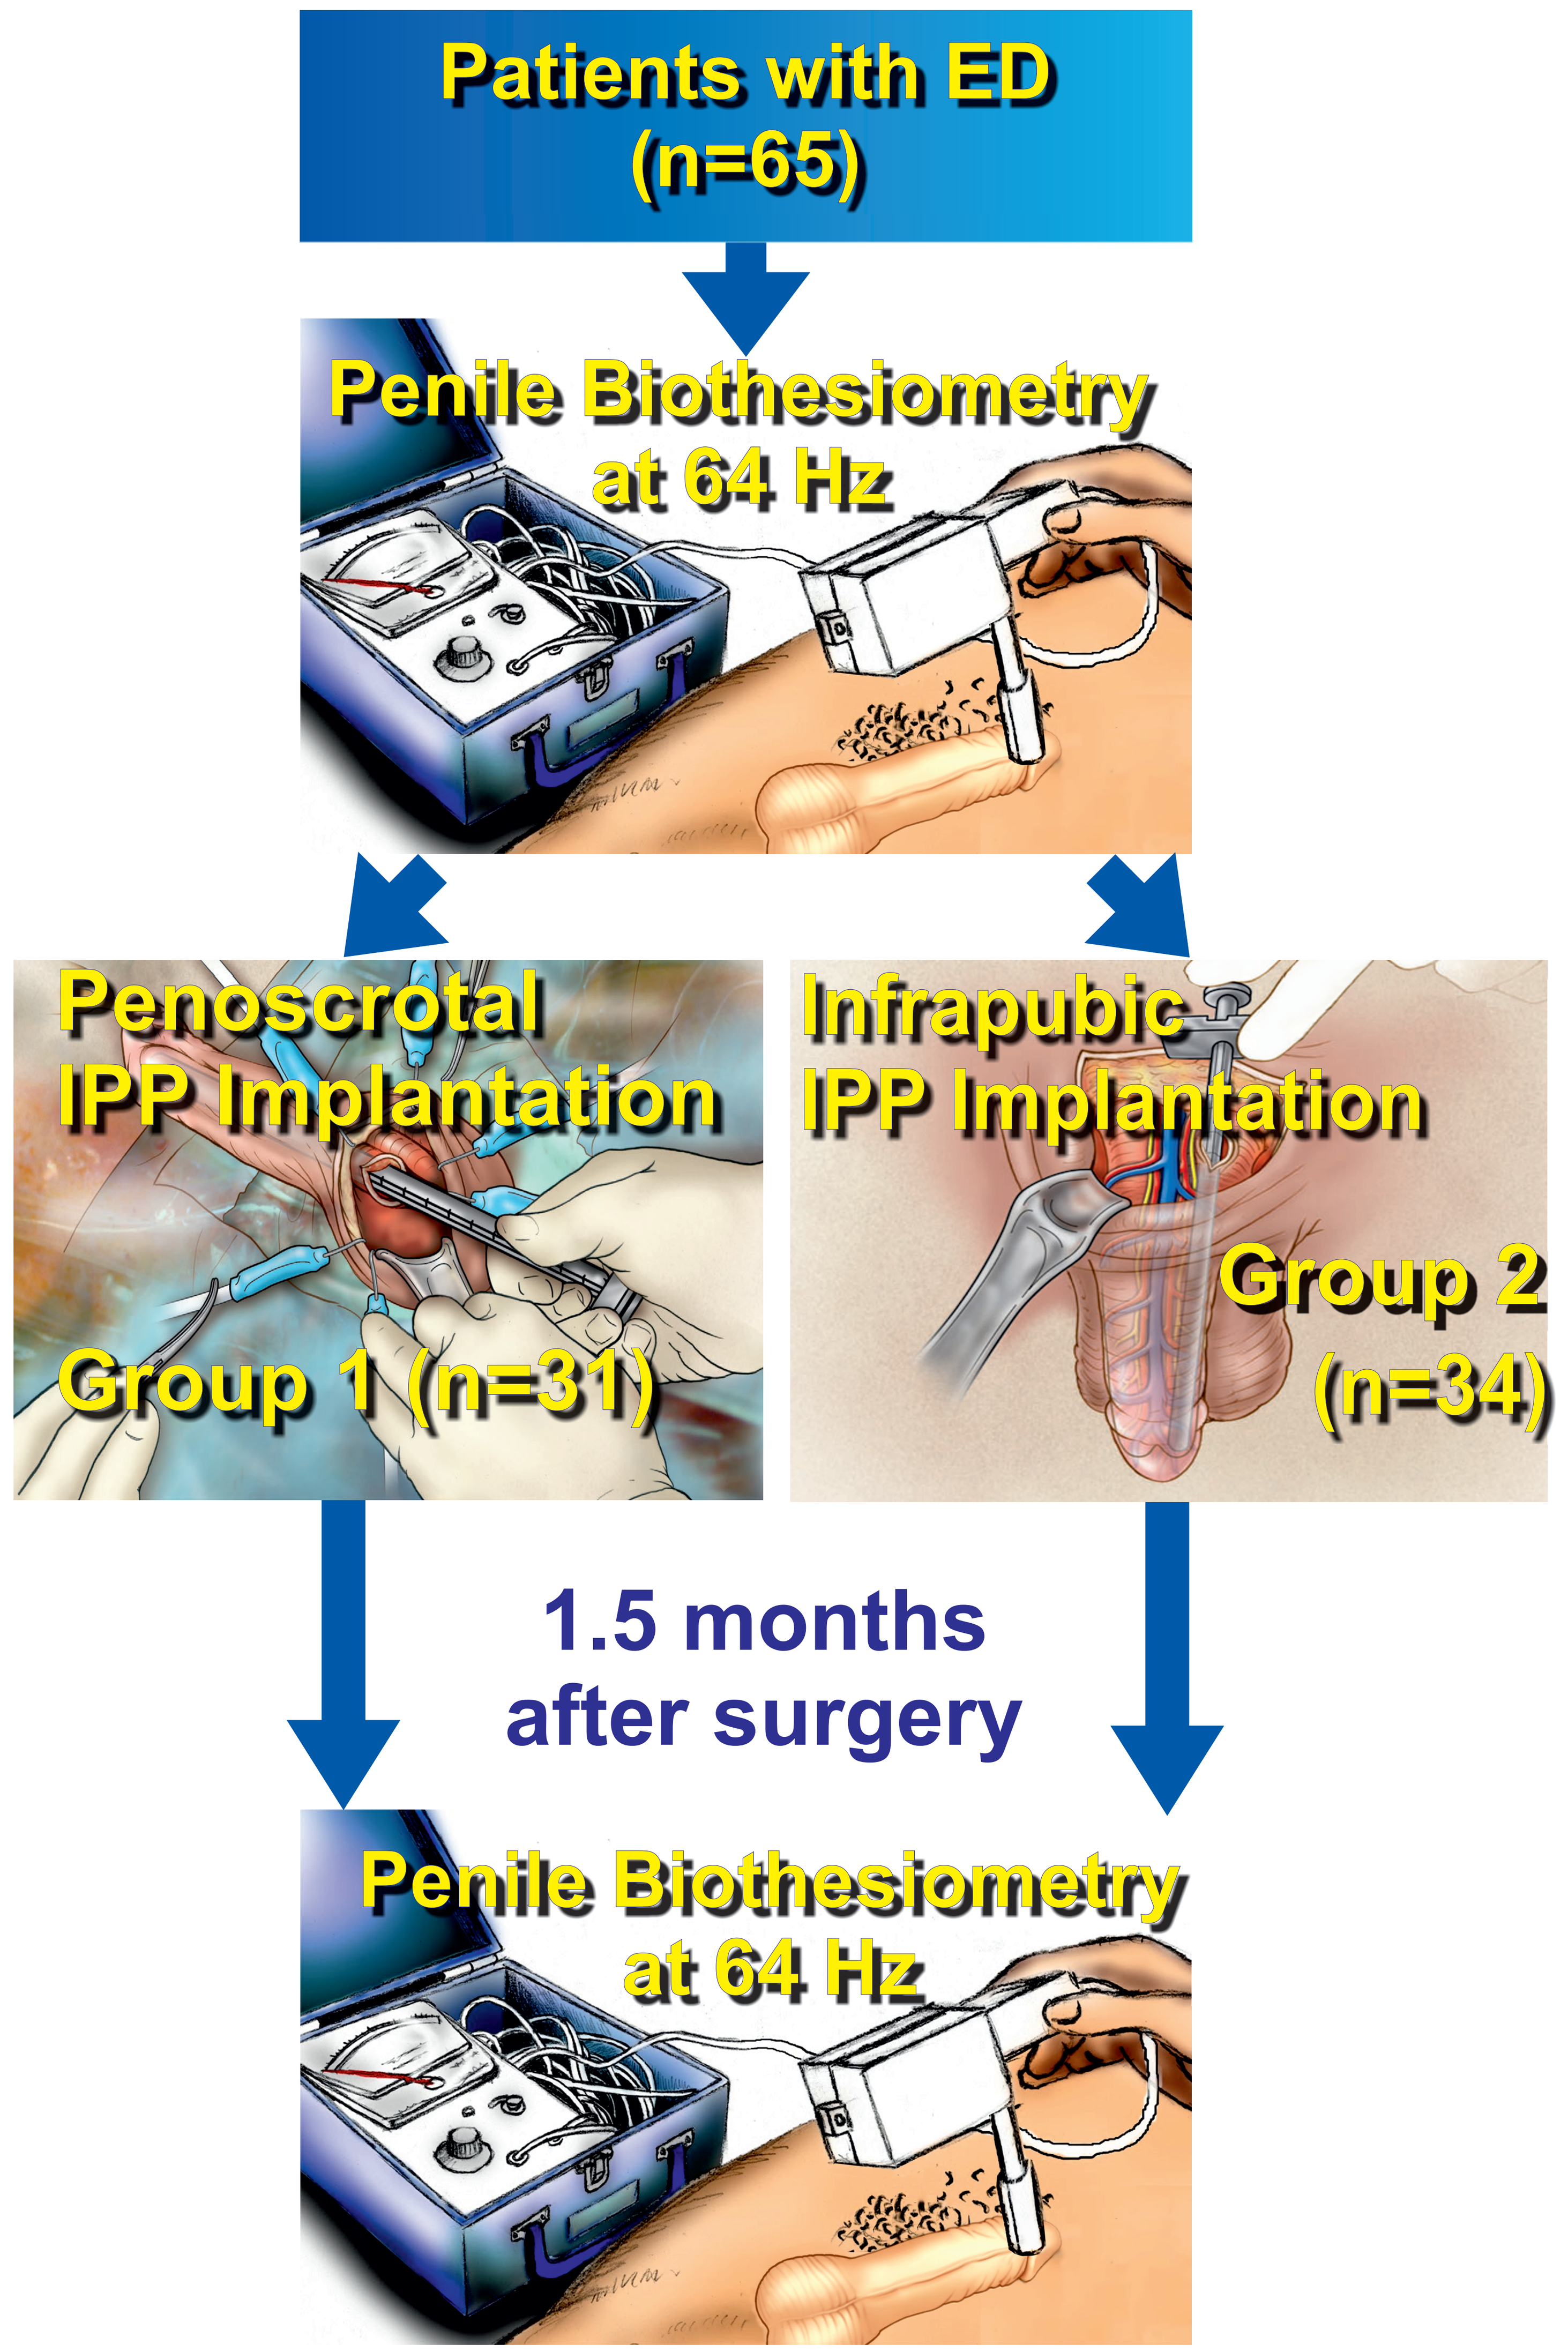

Patients with ED
(n=65)
Penile Biothesiometry
at 64 Hz
Penoscrotal
IPP Implantation
Group 1 (n=31)
Infrapubic
IPP Implantation
Group 2
(n=34)
1.5 months
after surgery
Penile Biothesiometry
at 64 Hz

RESULTS:

In group 1, PSV prior to and after surgery was 20.29±3.21 and 20.81±2.88 dB, respectively (p=0.484). In group 1, PSV prior to and after the procedure was 19.76±3.39 and 20.61±3.25 dB, respectively (p=0.303). No statistically significant differences in pre- and post-operative parameters between the groups were observed (p=0.914 and p=0.787).

PENILE SENSITIVITY TO VIBRATION BEFORE AND AFTER IPP IMPLANTATION VIA A PENOSCROTAL AND INFRAPUBIC APPROACH

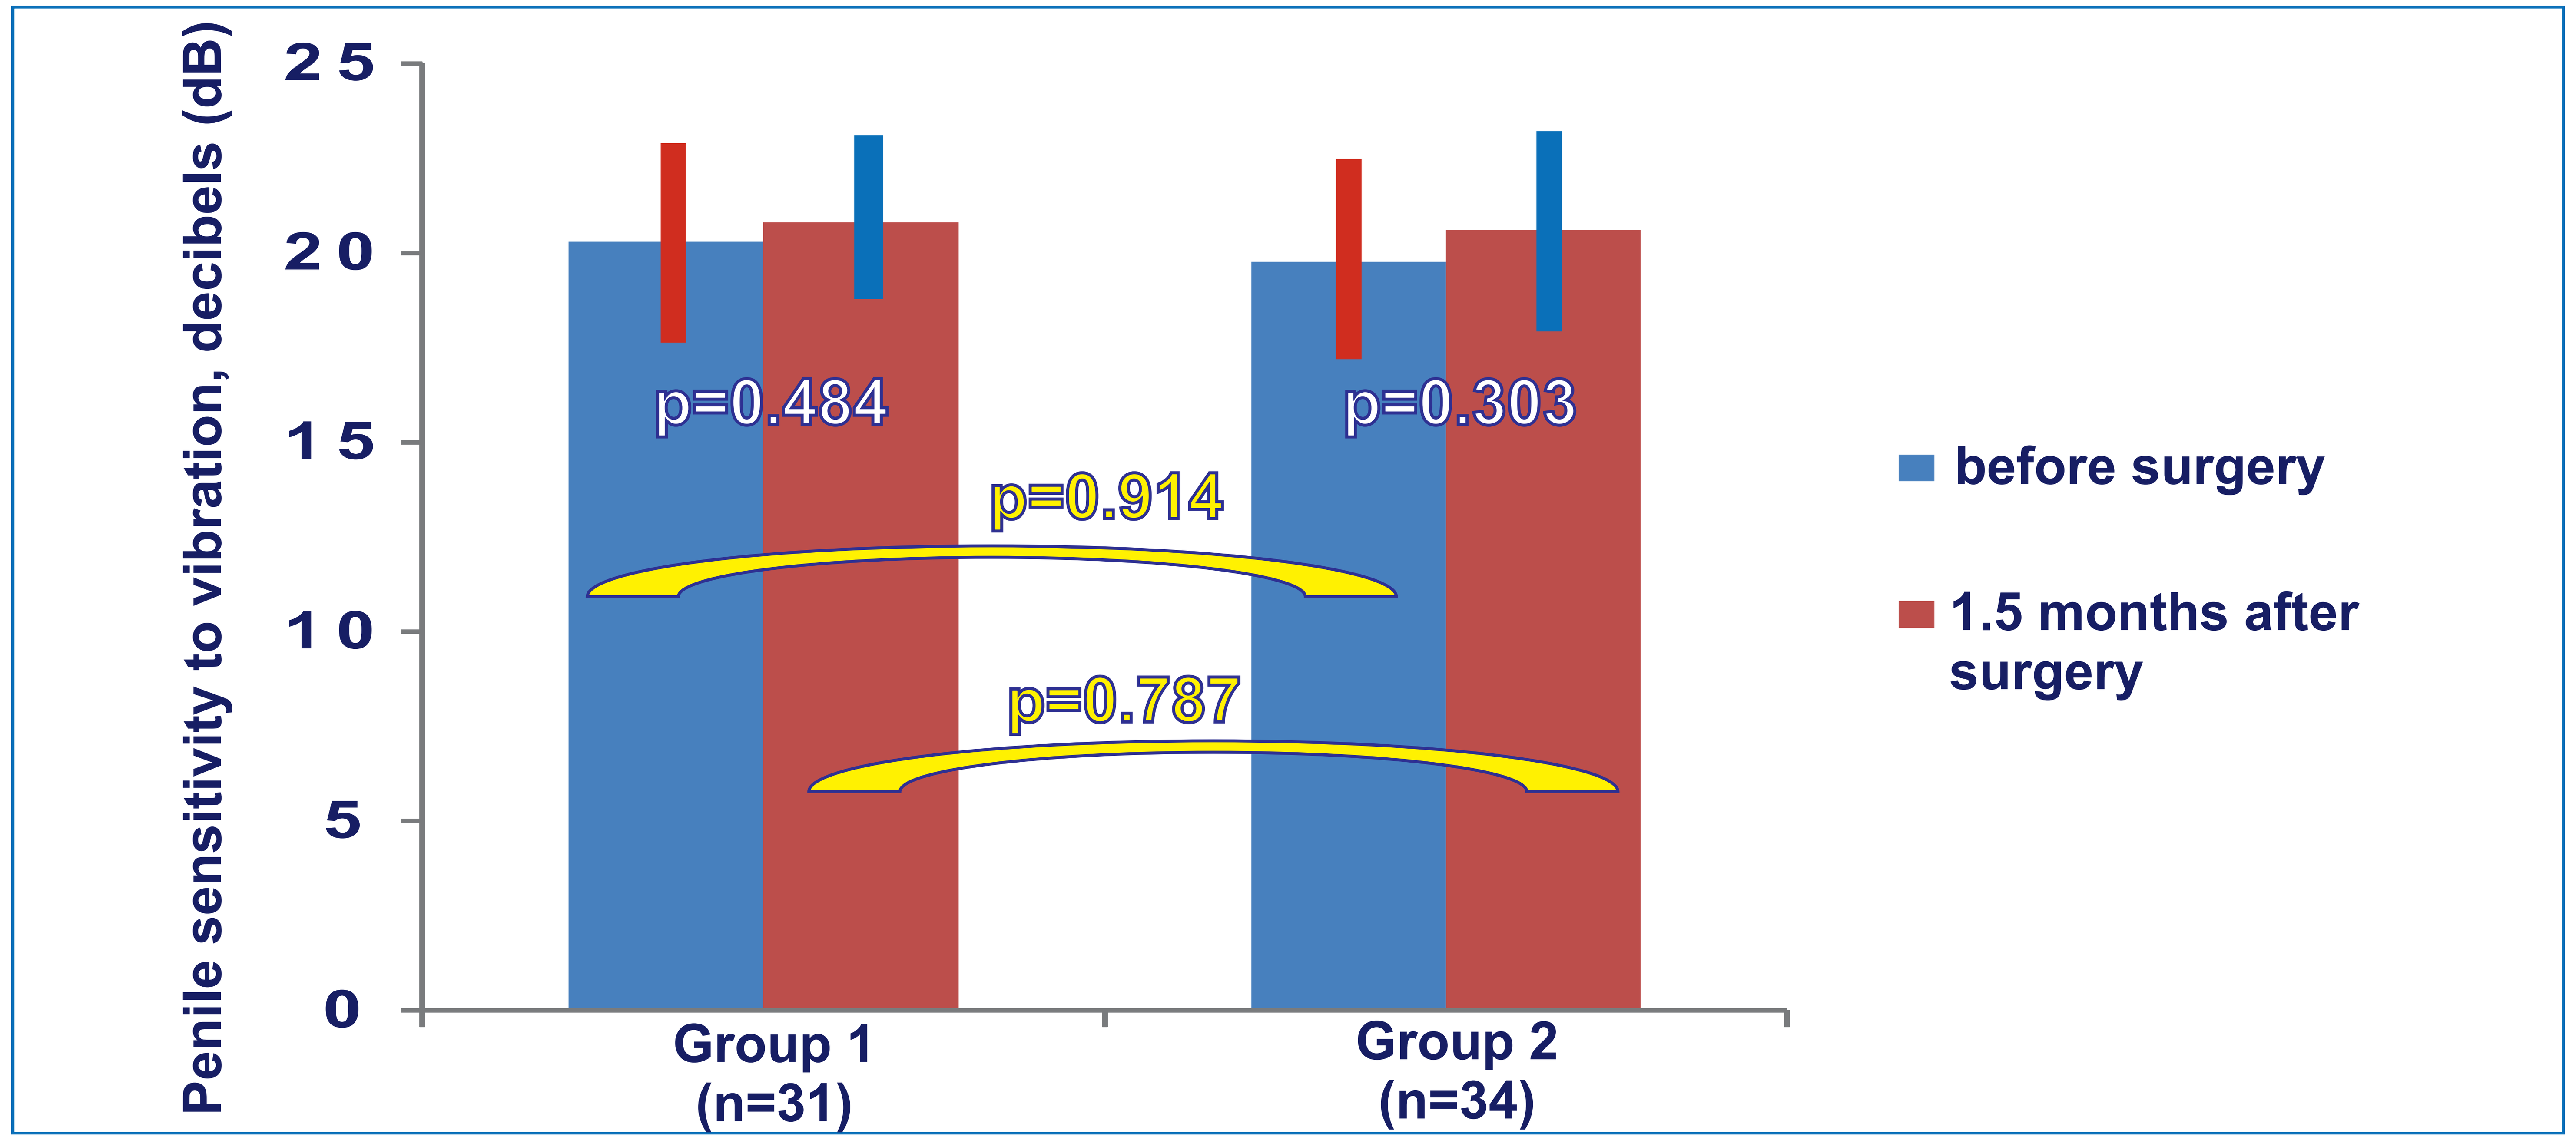

Penile sensitivity to vibration, decibels (dB)
25
20
15
10
5
0
p=0.484
p=0.303
p=0.914
p=0.787
Group 1
(n=31)
Group 2
(n=34)
before surgery
1.5 months after surgery

CONCLUSIONS:

IPP implantation using a penoscrotal or infrapubic approach is not associated with a statistically significant decrease in PSV. In order to avoid damaging dorsal nerve fibers when using a infrapubic approach, optical magnification should be used prior to corporotomy.